The Impact of Disease Severity on Responder Rates in a Phase 2b Study of XEN1101, a Potent, Selective Potassium Channel Opener, in Adults With Focal Epilepsy (X-TOLE)

Roger Porter,[1] Jacqueline French,[2] Emilio Perucca,[3] Martin Brodie,[4] Cynthia Harden,[5] Constanza Luzon Rosenblut,[5] Jenny Qian,[5] Christopher Kenney,[5] Gregory N. Beatch[5]

[1]University of Pennsylvania, Philadelphia, PA; [2]New York University Grossman School of Medicine and NYU Langone Health, New York, NY; [3]Monash University, Melbourne, Victoria, Australia, and University of Melbourne (Austin Health), Heidelberg, Victoria, Australia; [4]University of Glasgow Department of Medicine and Therapeutics, Western Infirmary, Glasgow, Scotland; [5]Xenon Pharmaceuticals Inc., Vancouver, BC, Canada

INTRODUCTION

- XEN1101 is a novel, potent K_V7 potassium channel opener in development for the treatment of focal onset seizures (FOS), primary generalized tonic-clonic seizures, and major depressive disorder[1-5]
- X-TOLE is a completed Phase 2b, randomized, double-blind, placebo-controlled, parallel-group, dose-ranging, multicenter study with an ongoing optional 5-year open-label extension (OLE) evaluating the efficacy, safety, and tolerability of XEN1101 administered with food as adjunctive treatment in adults with FOS[6]
- In X-TOLE, XEN1101 (10, 20, and 25 mg once daily [QD] with food) showed a dose-dependent, highly statistically significant, and rapid-onset reduction in FOS frequency[6,7]
- XEN1101 was generally well tolerated, with adverse events consistent with commonly prescribed antiseizure medications (ASMs)[6]
- Compared to other recent adult FOS clinical studies (**Table 1**), X-TOLE included a difficult-to-treat patient population given the baseline seizure burden, number of tried and stopped ASMs, and number of concomitant ASMs during the study[6]

Table 1. Summary of Recent FOS Trials: Baseline Seizure Frequencies and Concomitant ASMs

Drug	Phase (Study Years)	Total N (Population)	BL Median Monthly Seizure Frequency, Mean (SD)*	BL Median Monthly Seizure Frequency, Median (Min, Max)†	Allowed Concomitant ASMs	Concomitant ASMs ≤2, % of Patients	Concomitant ASMs = 3, % of Patients
XEN1101[6]	Phase 2b (2019–2021)	325 (Safety)	N/A‡	13.5 (13.5,13.5)	1 to 3	49.5%	50.5%
Cenobamate[8,9]	Phase 2 and 3 (2011–2015)	659 (Safety)	8.5 (1.9)	8.7 (5.5, 11)	1 to 3	70.1%	29.6%§
Brivaracetam[10-14]	Phase 2 and 3 (2005–2014)	1919 (ITT)	9.1 (1.3)	9.0 (7.0, 11.8)	1 to 2	96.1%	3.8%¶
Perampanel[15-18]	Phase 2 (2005–2007)	153 (Safety)	N/A#	N/A#	1 to 2	99.3%‖	0%
	Phase 2 and 3 (2007–2010)	1526 (Safety)	11.9 (1.8)	11.9 (9.3, 14.3)	1 to 3	64.8%	35.2%
Lacosamide[19-21]	Phase 2 (2002–2004)	415 (Safety)	N/A**	11-13‖	1 to 2	100%	0%
	Phase 2 and 3 (2004–2006)	879 (Safety)	12.5 (2.7)	11.5 (9.9, 16.5)	1 to 3	67.6%	32.4%

*ASM, antiseizure medication; BL, baseline; FOS, focal onset seizure. *Calculated as the mean (SD) of median monthly seizure frequency reported in each referenced study. †Calculated as the median (min, max) of median monthly seizure frequency reported in each referenced study. ‡Requires multiple studies for analysis. §Some additional patients received temporary treatment with a fourth ASM. ¶Subset of patients used benzodiazepines as needed. #Not reported. ‖As reported. **Not calculated due to method used to report baseline median monthly seizures.*

- The study population of X-TOLE included a spectrum of disease severity, allowing for comparison of the effect of baseline disease severity characteristics on efficacy outcomes
- A prior post hoc analysis of X-TOLE suggested that the efficacy of XEN1101 25 mg QD with food, measured by the median percentage change (MPC) in monthly FOS frequency, may be most pronounced in patients with less-severe disease[22]
- Another widely used efficacy endpoint for evaluating ASM efficacy is the percentage of patients with a ≥50% reduction in seizure frequency during a given treatment period compared with baseline, known as the responder rate or RR50. We report the potential impact of baseline disease severity on the RR50 in patients treated with XEN1101 25 mg QD in X-TOLE

METHODS

- Key eligibility criteria: aged 18–75 years with a diagnosis of focal epilepsy per International League Against Epilepsy criteria (≥2 years),[23] ≥4 countable focal seizures per month during a planned 8-week baseline period, and receiving stable treatment with 1 to 3 ASMs
- A total of 325 patients were randomized to an 8-week double-blind period (DBP) and treated across 3 active treatment groups or placebo in a 2:1:1:2 ratio (25 mg: 20 mg: 10 mg: placebo, QD with food; **Figure 1**)

Figure 1. Study Design

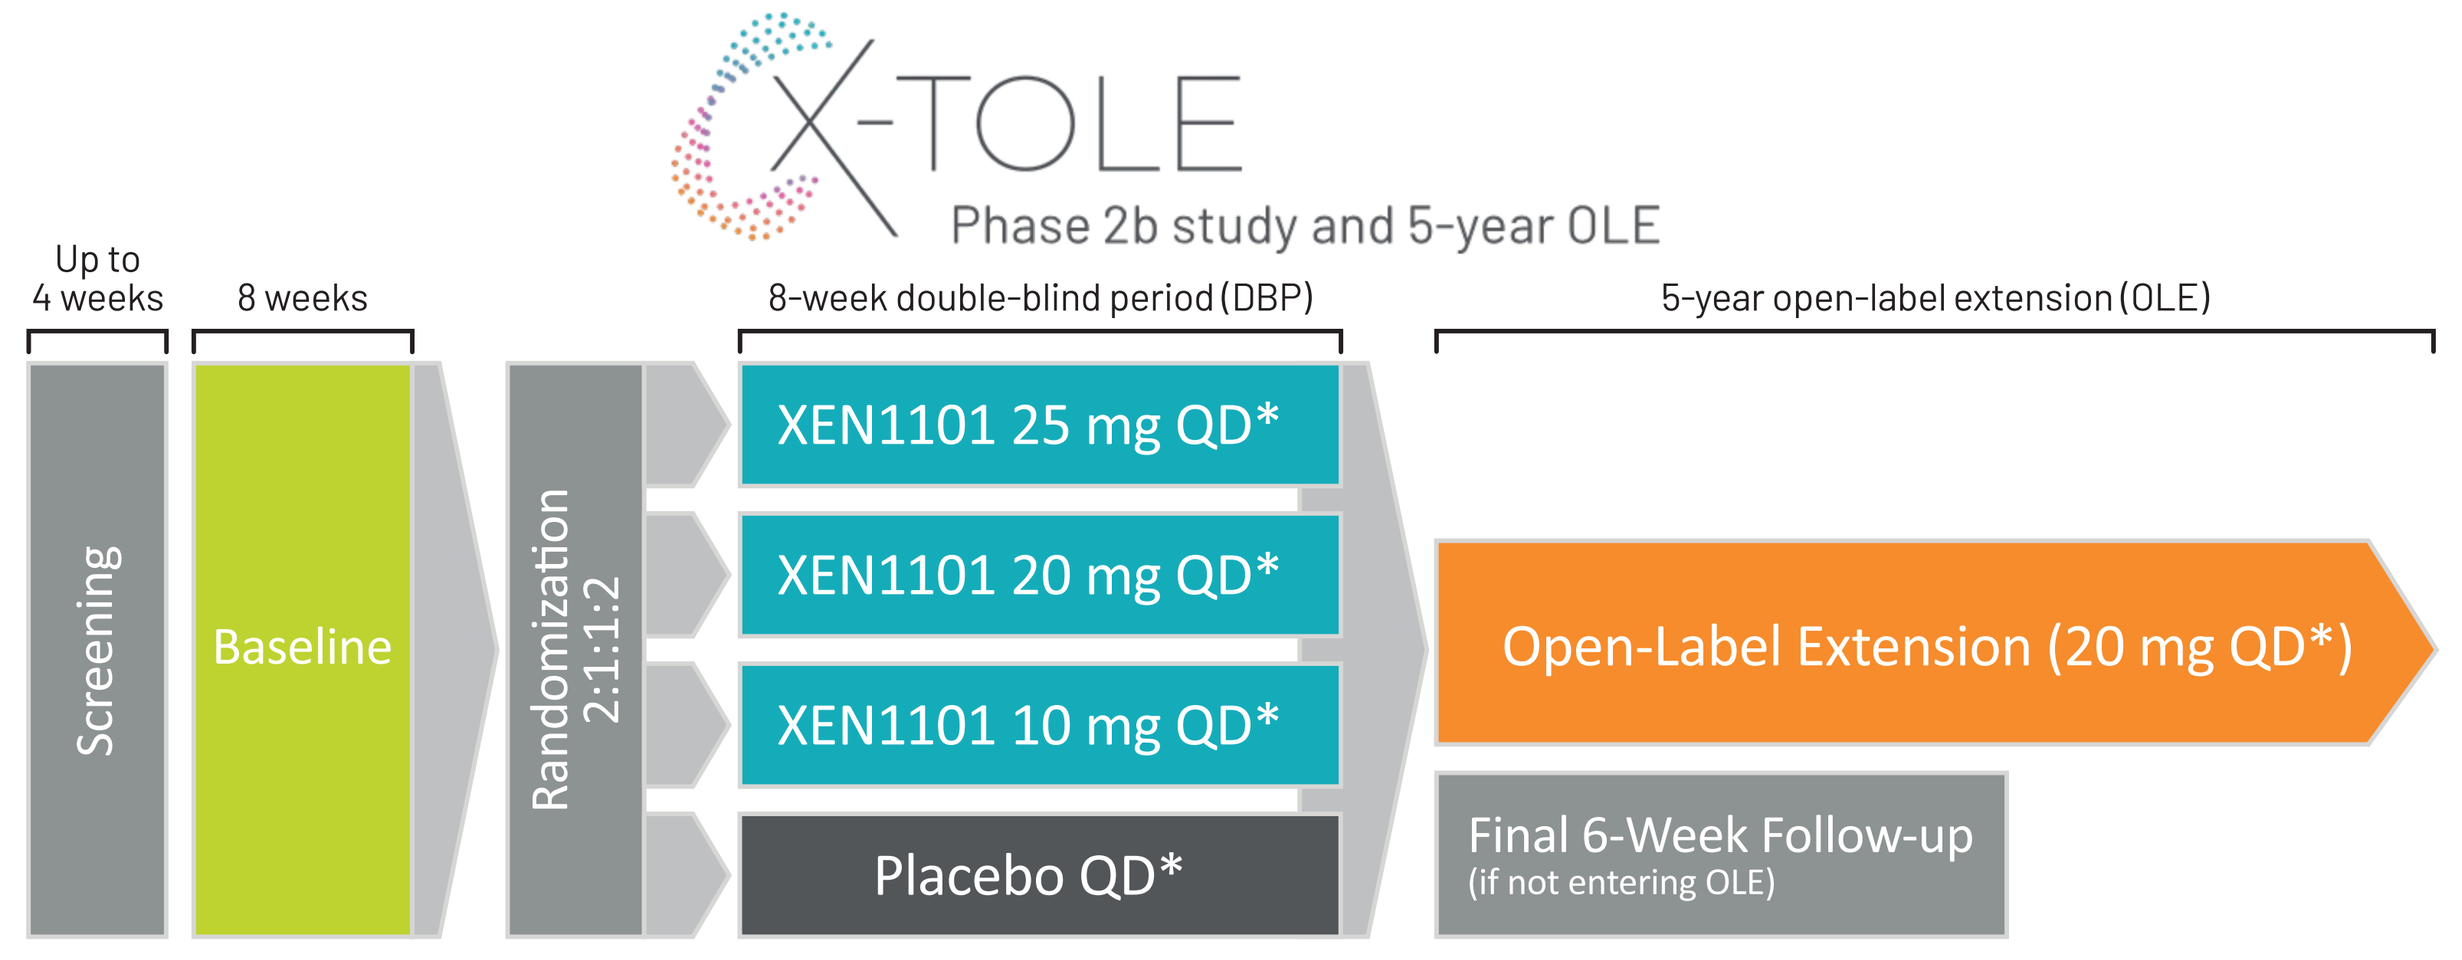
X-TOLE
Phase 2b study and 5-year OLE
Up to 4 weeks
8 weeks
8-week double-blind period (DBP)
5-year open-label extension (OLE)
Screening
Baseline
Randomization 2:1:1:2
XEN1101 25 mg QD*
XEN1101 20 mg QD*
XEN1101 10 mg QD*
Placebo QD*
Open-Label Extension (20 mg QD*)
Final 6-Week Follow-up
(if not entering OLE)

**Administered as a once-daily capsule with food with no titration required. QD, once daily.*

- Post hoc analyses were performed to assess the influence of baseline disease severity as indicated by seizure frequency (≤8.5 and >8.5 seizures per month), number of concomitant ASMs (≤2 or =3), and number of previously tried and stopped ASMs (median of ≤6 and >6) on the responder rate, defined as the RR50 in monthly FOS frequency from baseline to the DBP. The post hoc analysis data presented below pertain to patients given 25 mg QD with food (25 mg group)
 – The ≤8.5 and >8.5 seizures per month frequency was based on the average median baseline seizure frequencies of the cenobamate randomized controlled trials (**Table 1**)

RESULTS

Demographics and Baseline Characteristics

- At DBP baseline, patients in the 25 mg group (n=114) had a median of 12.8 monthly seizures and had tried and discontinued a median of 6 ASMs; 48.2% were taking 3 concomitant ASMs[6] (**Table 2**)
- Patients in the placebo group (n=114) had a median of 13.4 monthly seizures and had tried and discontinued a median of 6 ASMs at DBP baseline; 49.1% were taking 3 concomitant ASMs[6] (**Table 2**)

Table 2. Demographic and DBP Baseline Characteristics of the Placebo and 25 mg Groups[6]

	Placebo (n=114)	XEN1101 25 mg (n=114)
Age at study entry, mean (SD), y	42.9 (13.7)	38.7 (13.1)
≥65	5 (4.4)	1 (0.9)
<65	109 (95.6)	113 (99.1)
Sex, n (%)		
Female	61 (53.5)	54 (47.4)
Male	53 (46.5)	60 (52.6)
Region, n (%)		
Europe	67 (58.8)	68 (59.6)
North America	47 (41.2)	46 (40.4)
Baseline seizure frequency per mo		
Mean (SD)	27.3 (38.5)	23.5 (30.4)
Median	13.4	12.8
Background ASM use, n (%)		
1 ASM	12 (10.5)	11 (9.6)
2 ASMs	46 (40.4)	48 (42.1)
3 ASMs	56 (49.1)	55 (48.2)
Number of prestudy ASMs failed, n (%)		
≤3 ASMs	29 (25.4)	31 (27.2)
>3 ASMs,	85 (74.6)	83 (72.8)
Median (IQR)	6.0 (4.0–8.0)	6.0 (3.0–9.0)

ASM, antiseizure medication; IQR, interquartile range.

Efficacy Results: Baseline Disease Severity Impact on RR50 in the 25 mg Group

Baseline Seizure Sub-Group Analysis

- RR50 was achieved by 65.5% of patients with ≤8.5 seizures per month at baseline and 50.6% with >8.5 seizures per month; with placebo treatment, RR50 was higher (20.0% vs 13.1%) in the ≤8.5 vs >8.5 seizure per month sub-group (**Figure 2**)

Number of Tried and Stopped ASMs Sub-Group Analysis

- RR50 was achieved by 64.2% of patients with ≤6 tried and stopped ASMs and 40.0% with >6 ASMs; with placebo treatment, RR50 was higher (19.4% vs 8.5%) in the ≤6 vs >6 tried and stopped ASMs sub-group (**Figure 3**)

Concomitant ASMs Sub-Group Analysis

- RR50 was achieved by 56.9% of patients treated with 1–2 concomitant ASMs throughout and 51.9% treated with 3 concomitant ASMs throughout the study (**Figure 4**)

Figure 2. Baseline Seizure Sub-Group Analysis

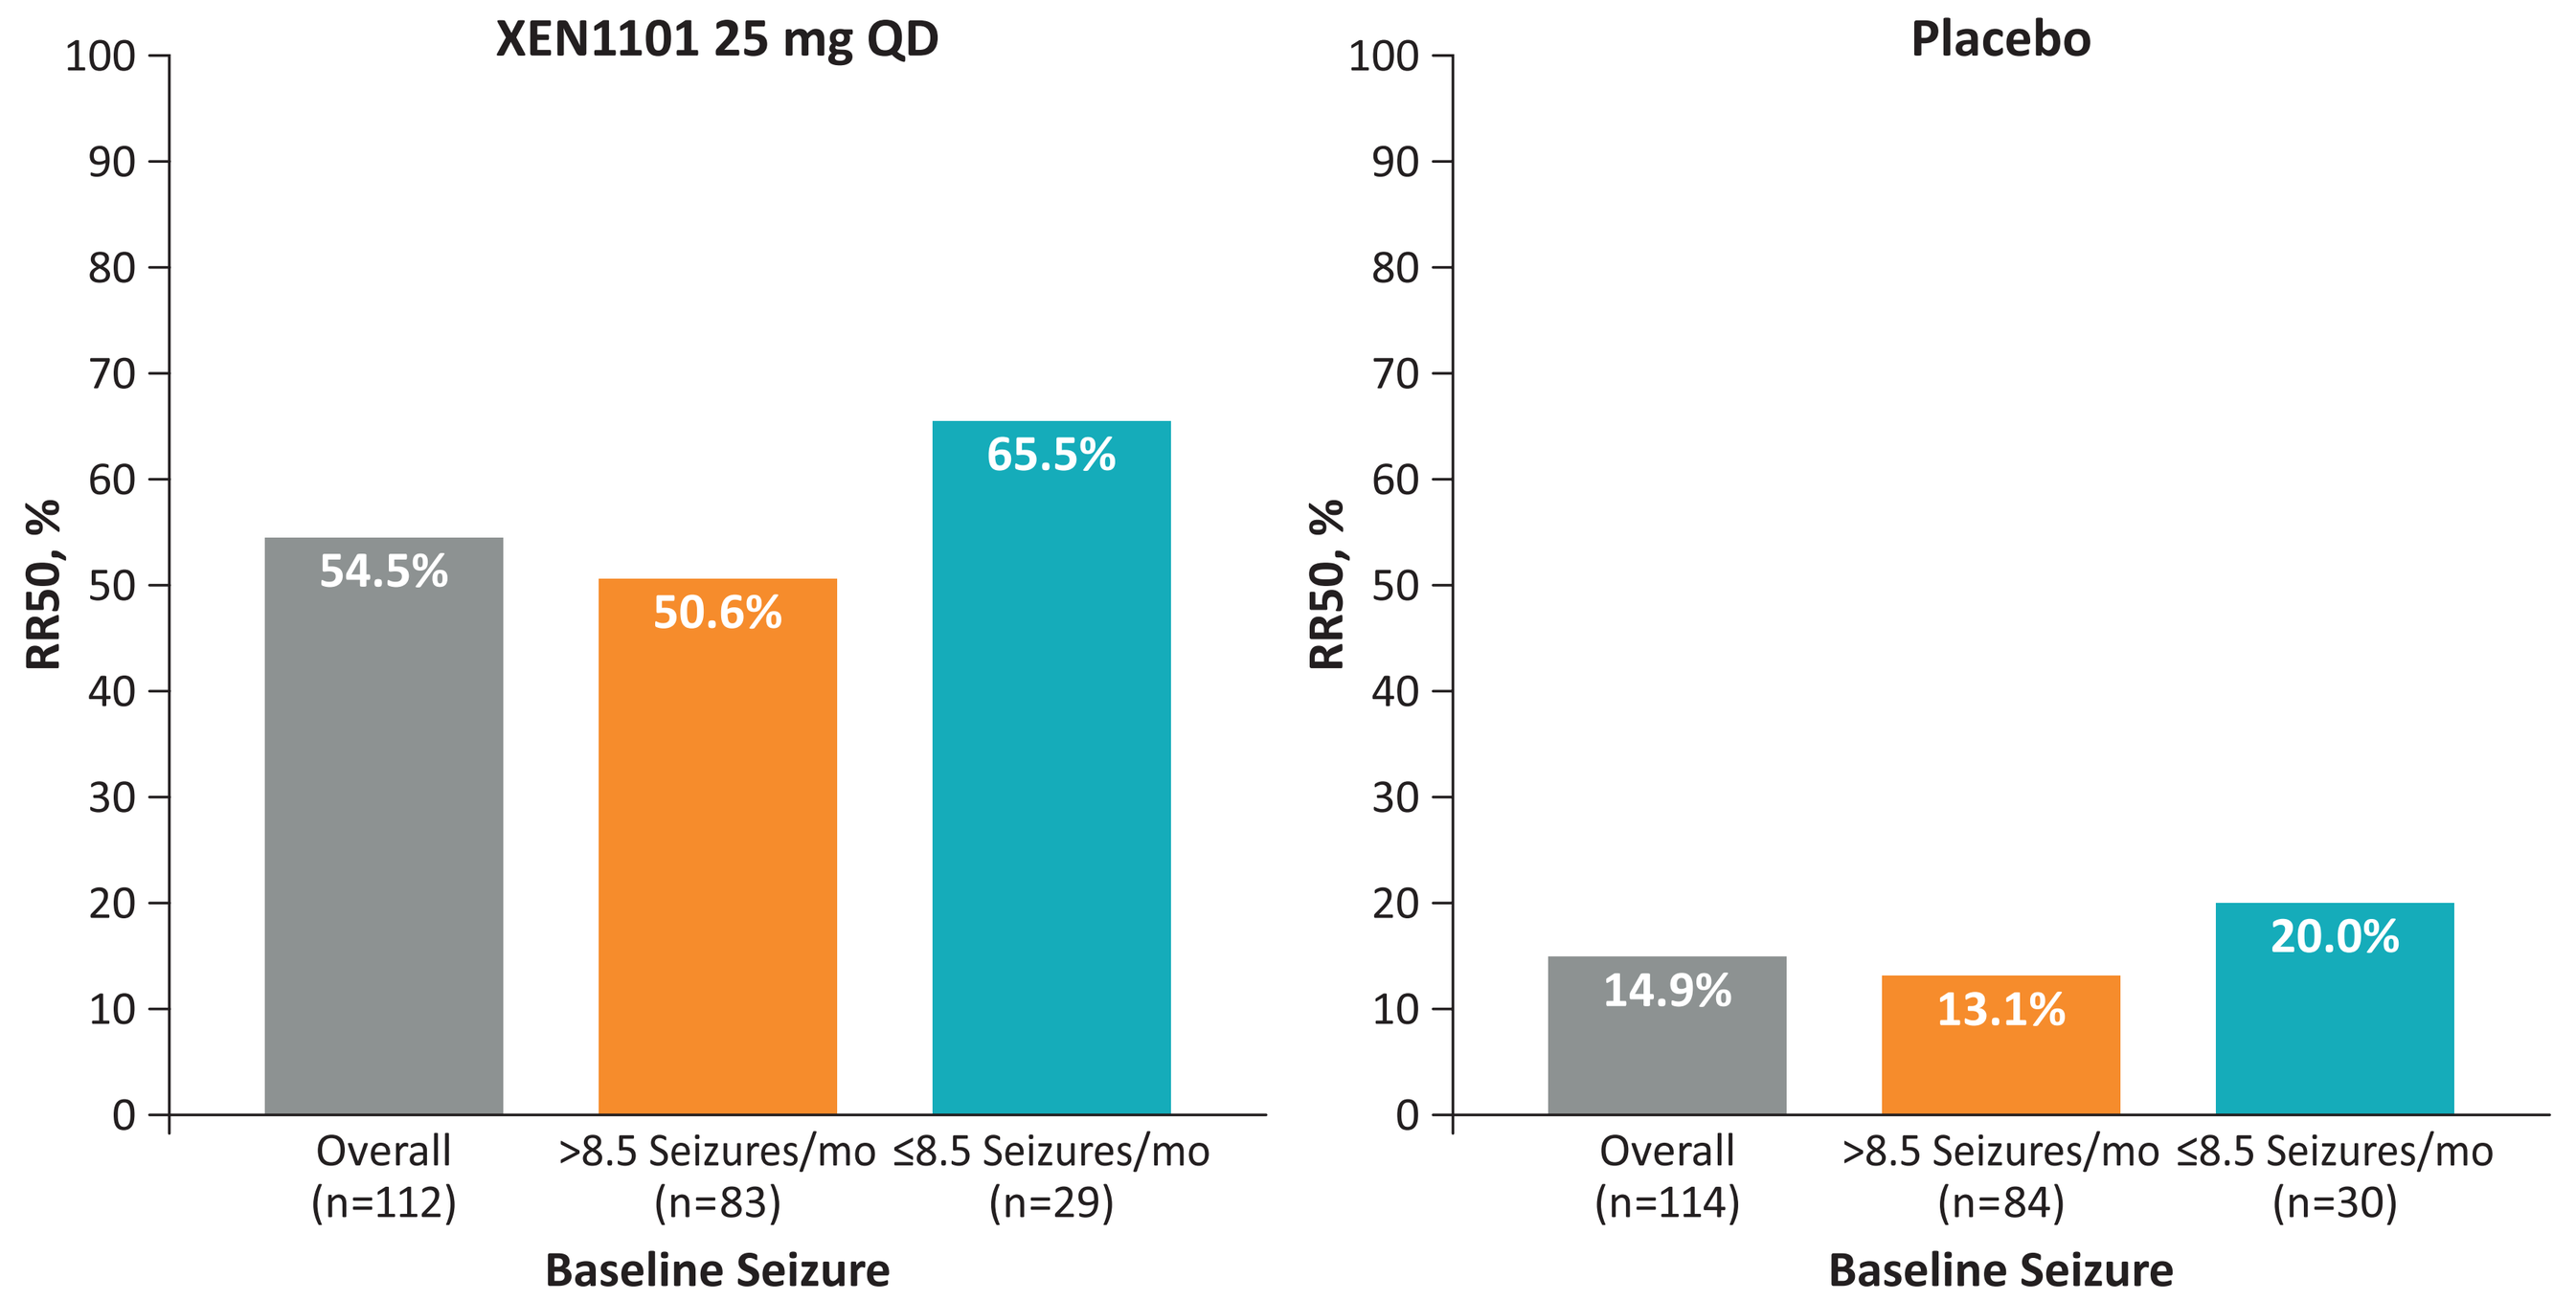
XEN1101 25 mg QD
Placebo
RR50, %
54.5%
50.6%
65.5%
14.9%
13.1%
20.0%
Overall (n=112)
>8.5 Seizures/mo (n=83)
≤8.5 Seizures/mo (n=29)
Overall (n=114)
>8.5 Seizures/mo (n=84)
≤8.5 Seizures/mo (n=30)
Baseline Seizure

Note: All doses taken with food. RR50, percentage of patients with a ≥50% reduction in seizure frequency.

Figure 3. Tried and Stopped Sub-Group Analysis

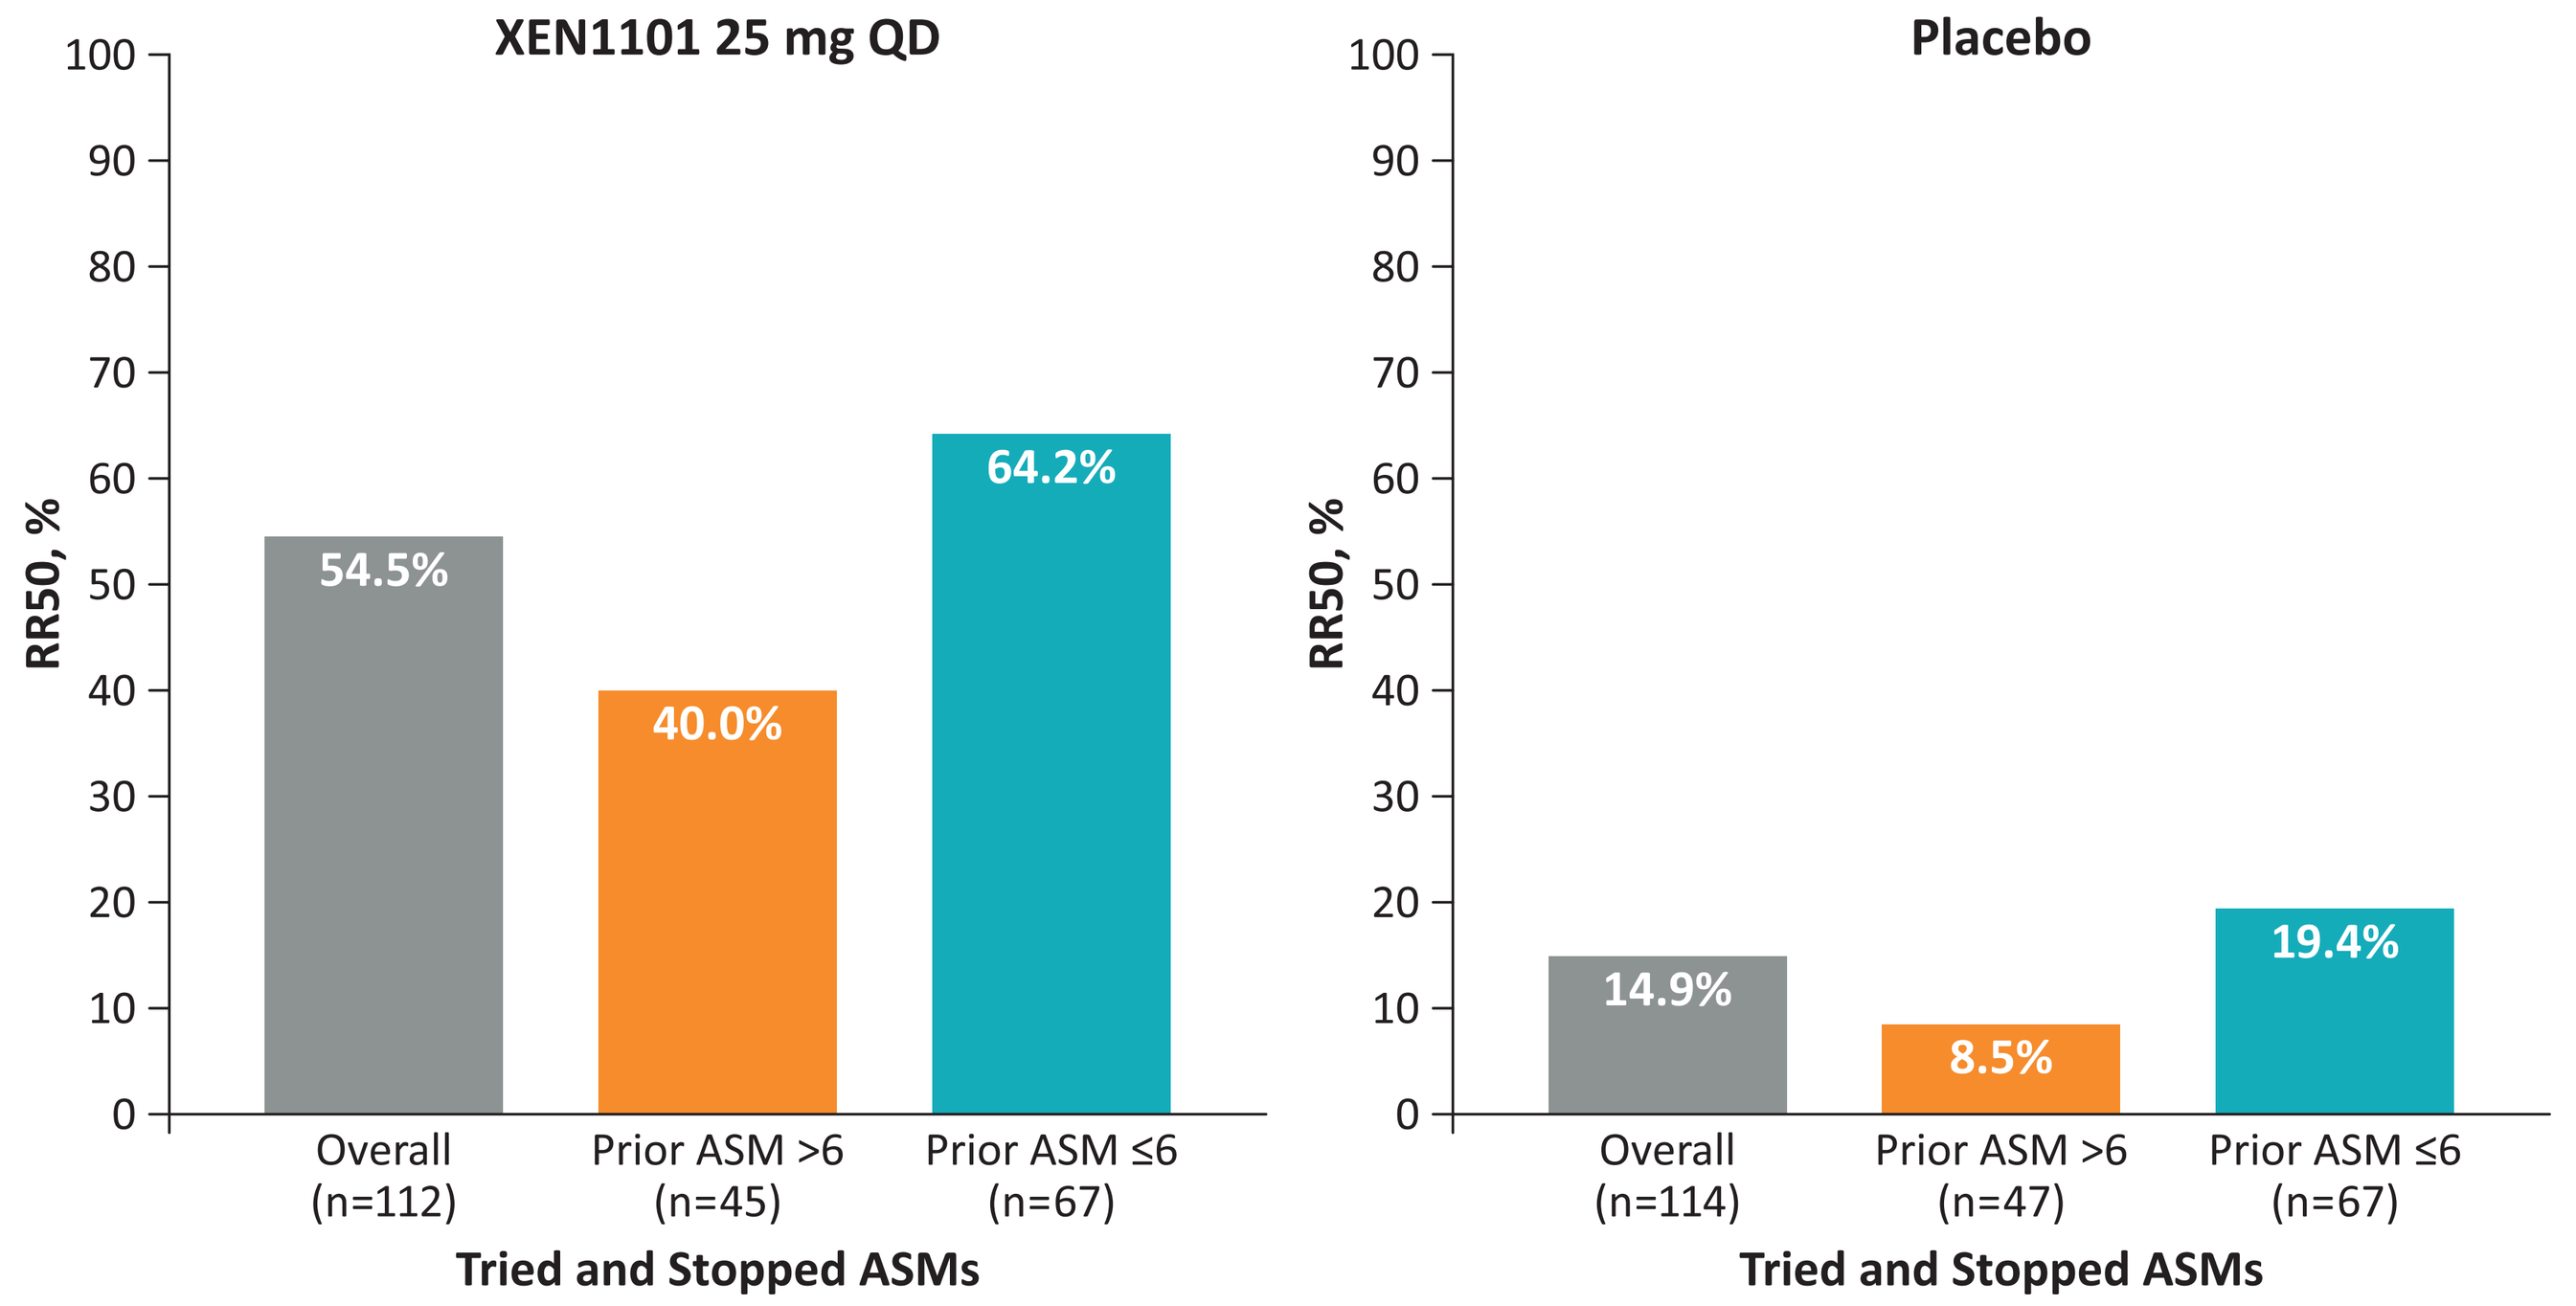
XEN1101 25 mg QD
Placebo
RR50, %
54.5%
40.0%
64.2%
14.9%
8.5%
19.4%
Overall (n=112)
Prior ASM >6 (n=45)
Prior ASM ≤6 (n=67)
Overall (n=114)
Prior ASM >6 (n=47)
Prior ASM ≤6 (n=67)
Tried and Stopped ASMs

Note: All doses taken with food. ASM, antiseizure medication; RR50, percentage of patients with a ≥50% reduction in seizure frequency.

Figure 4. Concomitant ASMs Sub-Group Analysis

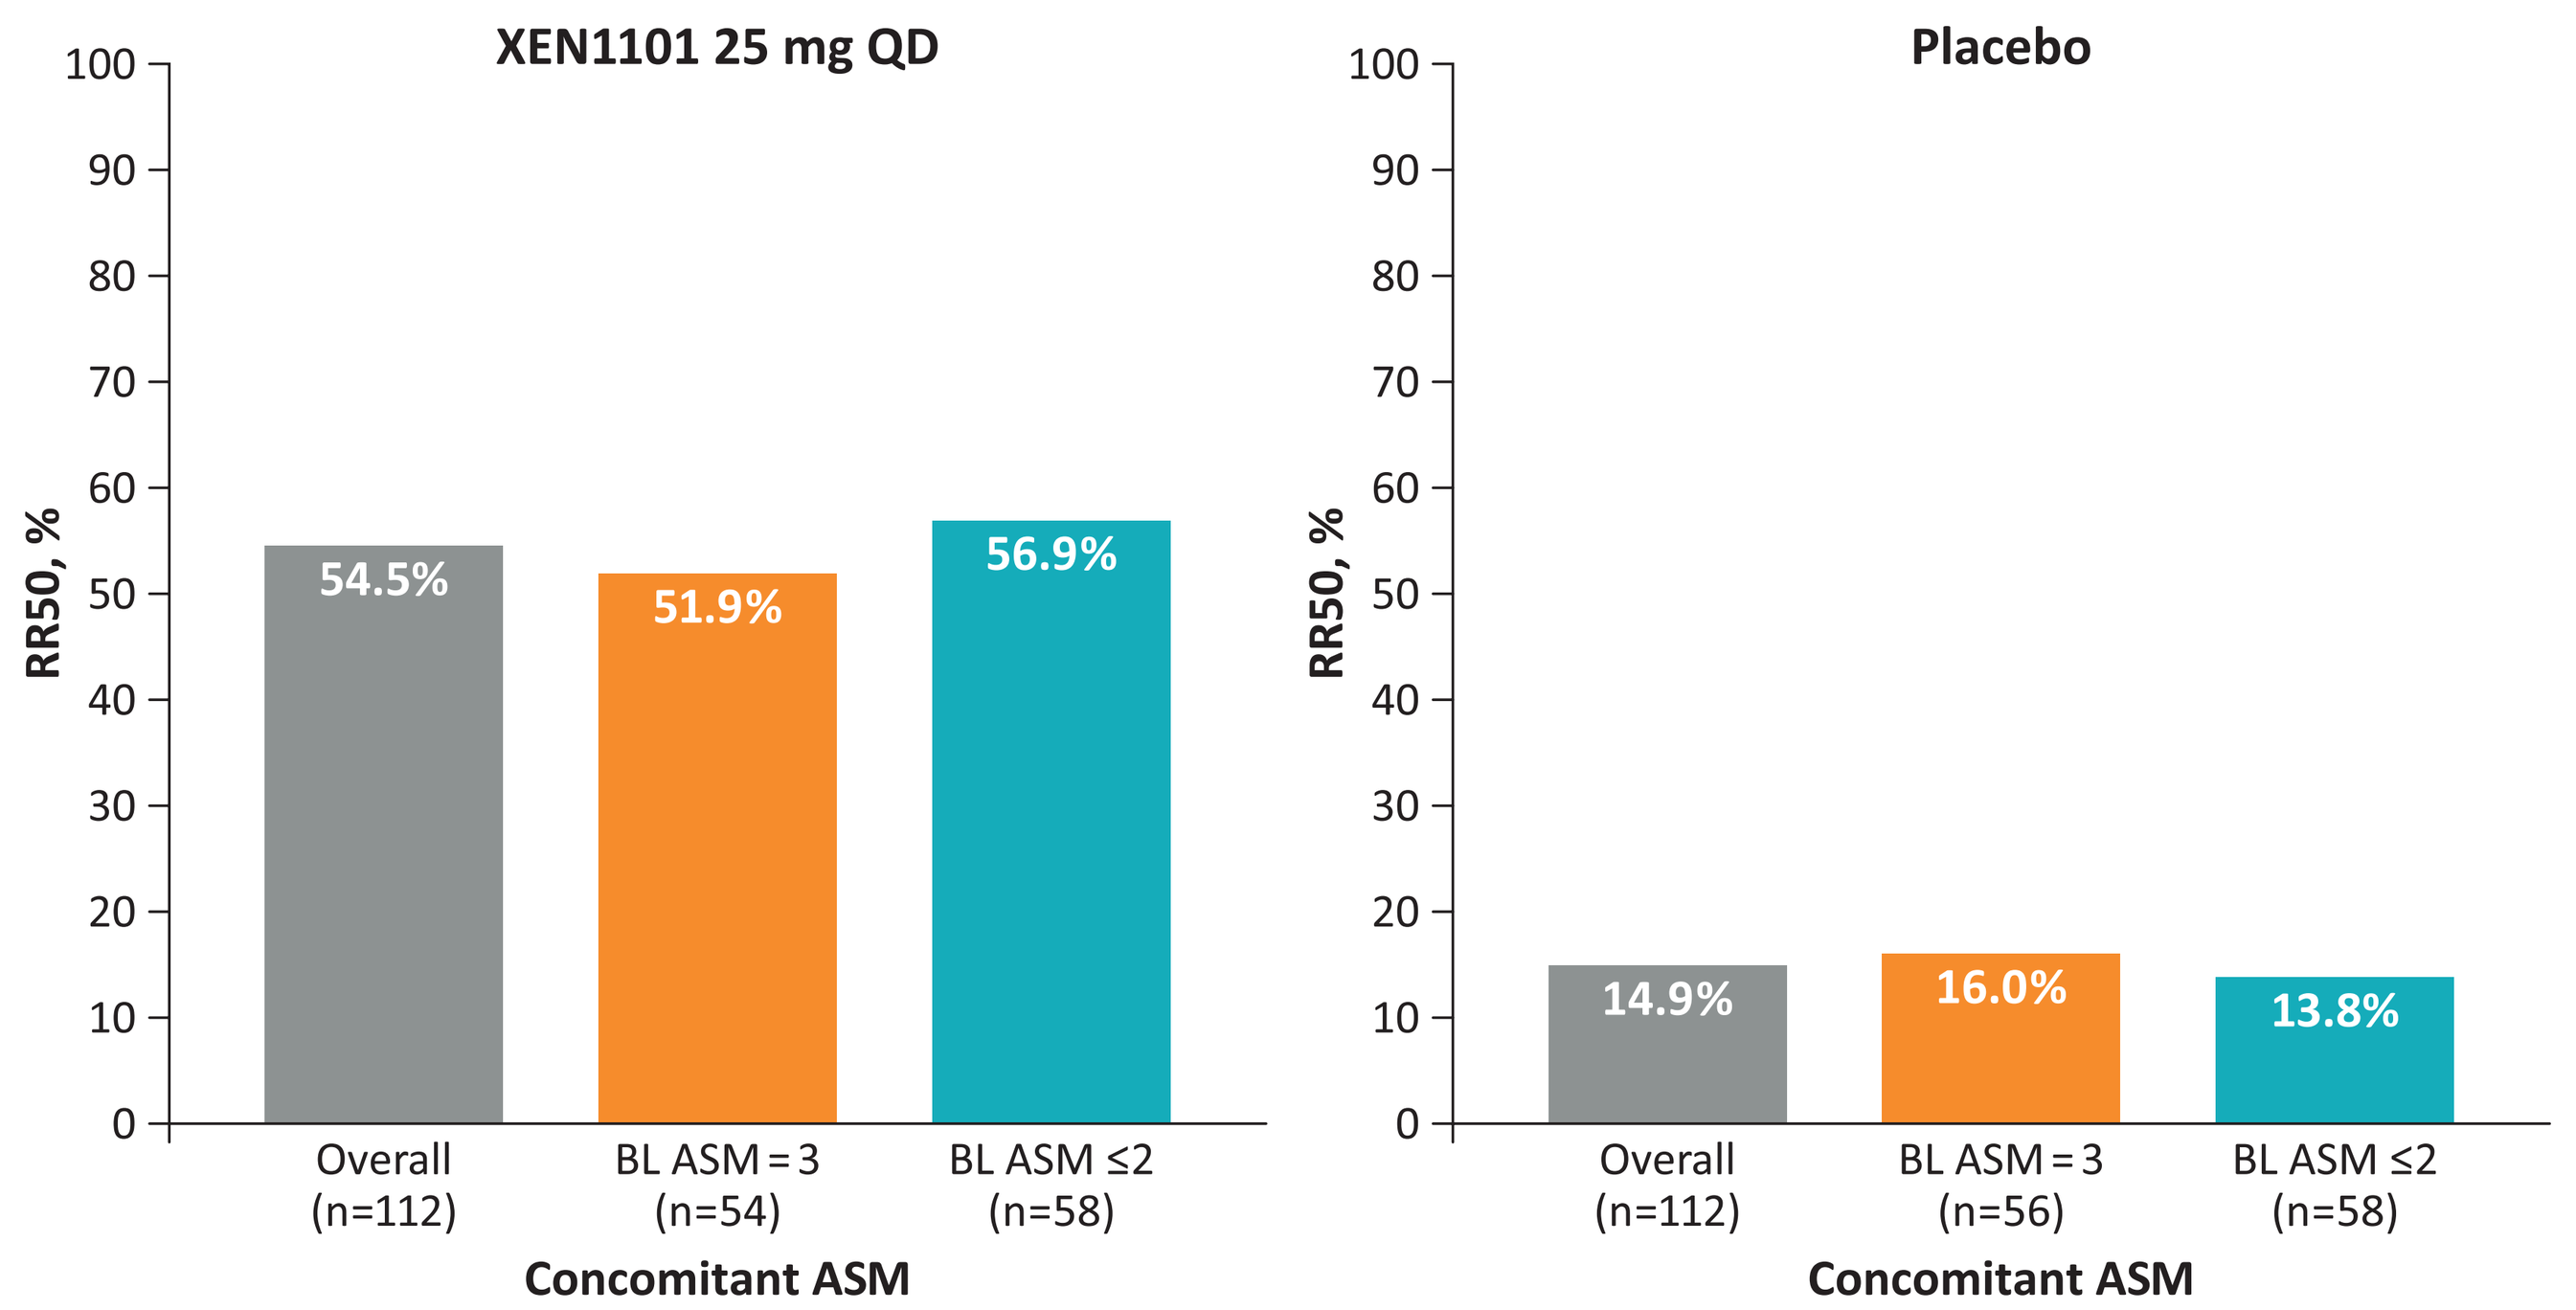
XEN1101 25 mg QD
Placebo
RR50, %
54.5%
51.9%
56.9%
14.9%
16.0%
13.8%
Overall (n=112)
BL ASM = 3 (n=54)
BL ASM ≤2 (n=58)
Overall (n=112)
BL ASM = 3 (n=56)
BL ASM ≤2 (n=58)
Concomitant ASM

Note: All doses taken with food. ASM, antiseizure medication; BL, baseline; RR50, percentage of patients with a ≥50% reduction in seizure frequency.

CONCLUSIONS

- Consistent with the significant MPC reduction in X-TOLE, 54.5% of the patients in the 25 mg group achieved the benchmark of RR50
- This effect was observed in a difficult-to-treat patient population
- XEN1101 was relatively more effective in patients with indicators of less-severe disease in the trial population
- These findings suggest that XEN1101 may be appropriate for use in patients with focal epilepsy across the spectrum of disease severity

ACKNOWLEDGMENTS Medical writing support was provided by Robin Smith, PhD, of The Curry Rockefeller Group, LLC (Tarrytown, NY), and was funded by Xenon Pharmaceuticals Inc.

FUNDING Xenon Pharmaceuticals Inc.

DISCLOSURES Roger Porter has been a consultant for Aeterna, Axonis, Cadent, Engrail, Longboard, Neurocrine, Otsuka, Passage Bio, and Xenon Pharmaceuticals Inc. Jacqueline French has numerous relationships on behalf of the Epilepsy Study Consortium with various commercial and academic entities (consulting, salary support, research support, travel reimbursement, or served on the editorial board), including Xenon Pharmaceuticals Inc. She receives salary support from the Epilepsy Study Consortium and no other income from these relationships. Emilio Perucca has received speaker or consultancy fees from Angelini, Arvelle, Biogen, Eisai, GW Pharma, Janssen, PMI Life Sciences, Sanofi, Shackelford Pharma, Sintetica, SK Life Science, Sun Pharma, Takeda, UCB Pharma, Xenon Pharmaceuticals Inc., and Zogenix. Martin Brodie has nothing to declare. Cynthia Harden, Constanza Luzon Rosenblut, Jenny Qian, Christopher Kenney, and Gregory N. Beatch are employees of and own stock or stock options in Xenon Pharmaceuticals Inc.

REFERENCES **1.** ClinicalTrials.gov. A Study to Evaluate XEN1101 as Adjunctive Therapy in Focal Epilepsy (X-TOLE). https://clinicaltrials.gov/study/NCT03796962 **2.** ClinicalTrials.gov. A Study to Evaluate XEN1101 as Adjunctive Therapy in Primary Generalized Tonic-Clonic Seizures (X-ACKT). https://clinicaltrials.gov/ct2/show/NCT05667142 **3.** ClinicalTrials.gov. A Randomized Study of XEN1101 Versus Placebo in Focal-Onset Seizures (X-TOLE3). https://clinicaltrials.gov/study/NCT05716100 **4.** ClinicalTrials.gov. A Study to Evaluate the Safety, Tolerability and Efficacy of XEN1101 in Major Depressive Disorder (X-NOVA). https://clinicaltrials.gov/study/NCT05376150 **5.** ClinicalTrials.gov. A Randomized Study of XEN1101 Versus Placebo in Focal-Onset Seizures (X-TOLE2). https://clinicaltrials.gov/study/NCT05614063 **6.** French JA, et al. *JAMA Neurol.* 2023;doi:10.1001/jamaneurol.2023.3542 **7.** Kenney C, et al. Rapid onset of efficacy of XEN1101, a novel potassium channel opener, in adults with focal epilepsy: results from a phase 2b study (X-TOLE) [Abstract number: 2.236]. 2022. https://cms.aesnet.org/abstractslisting/rapid-onset-of-efficacy-of-xen1101--a-novel-potassium-channel-opener--in-adults-with-focal-epilepsy--results-from-a-phase-2b-study-(x-tole) **8.** Chung SS, et al. *Neurology.* 2020;94(22):e2311-e2322. **9.** Krauss GL, et al. *Lancet Neurol.* 2020;19(1):38-48. **10.** Biton V, et al. *Epilepsia.* 2014;55(1):57-66. **11.** Klein P, et al. *Epilepsia.* 2015;56(12):1890-1898. **12.** French JA, et al. *Neurology.* 2010;75(6):519-525. **13.** Ryvlin P, et al. *Epilepsia.* 2014;55(1):47-56. **14.** Van Paesschen W, et al. *Epilepsia.* 2013;54(1):89-97. **15.** French JA, et al. *Neurology.* 2012;79(6):589-596. **16.** French JA, et al. *Epilepsia.* 2013;54(1):117-125. **17.** Krauss GL, et al. *Acta Neurol Scand.* 2012;125(1):8-15. **18.** Krauss GL, et al. *Neurology.* 2012;78(18):1408-1415. **19.** Ben-Menachem E, et al. *Epilepsia.* 2007;48(7):1308-1317. **20.** Chung S, et al. *Epilepsia.* 2010;51(6):958-967. **21.** Halasz P, et al. *Epilepsia.* 2009;50(3):443-453. **22.** Leung J, et al. The impact of disease severity on efficacy from a phase 2b study of XEN1101, a novel potassium channel opener, in adults with focal epilepsy (X-TOLE) [Abstract number: 2.233]. 2022. https://aesnet.org/abstractslisting/the-impact-of-disease-severity-on-efficacy-from-a-phase-2b-study-of-xen1101--a-novel-potassium-channel-opener--in-adults-with-focal-epilepsy-(x-tole) **23.** Fisher RS, et al. *Epilepsia.* 2017;58(4):522-530.

Poster 2.259 | American Epilepsy Society Annual Meeting | December 1–5, 2023 | Orlando, FL

XENON