XEN1101, a Novel Potassium Channel Modulator: Interim Data From an Ongoing, Long-Term, Open-Label Extension of a Phase 2B Study (X-TOLE) in Adults With Focal Epilepsy

Jacqueline French,[1] Roger Porter,[2] Emilio Perucca,[3] Martin Brodie,[4] Michael A. Rogawski,[5] Cynthia Harden,[6] Jenny Qian,[6] Constanza Luzon Rosenblut,[6] Christopher Kenney,[6] Gregory N. Beatch[6]

[1]New York University Comprehensive Epilepsy Center, New York, NY, USA; [2]University of Pennsylvania, Philadelphia, PA, USA; [3]University of Melbourne and Monash University, Melbourne, VIC, Australia; [4]University Department of Medicine and Therapeutics, Western Infirmary, Glasgow, Scotland, UK; [5]School of Medicine, University of California, Davis, Sacramento, CA, USA; [6]Xenon Pharmaceuticals Inc., Burnaby, BC, Canada

RATIONALE

- Despite the availability of several antiseizure medications (ASMs), 37% of patients with focal onset seizures (FOS) do not achieve ≥1 year of seizure freedom after a trial of 2 ASMs; the incremental likelihood of achieving seizure control decreases with each subsequent ASM[1]
- XEN1101 is a novel, potent, small-molecule, selective *KCNQ2/3* (K_v7.2/7.3) potassium channel opener in development for the treatment of FOS, primary generalized tonic-clonic seizures, and major depressive disorder[2]
- X-TOLE is a phase 2b, randomized, double-blind, placebo-controlled, parallel-group, dose-ranging, multicenter study with an optional 5-year open-label extension (OLE) evaluating the efficacy, safety, and tolerability of XEN1101 administered with food as adjunctive treatment in adults with FOS
 - In the double-blind period (DBP), XEN1101 treatment yielded a dose-dependent, consistent, highly statistically significant reduction in FOS across endpoints in a hard-to-treat patient population[2]
 - XEN1101 was generally well tolerated with a low incidence of serious adverse events (SAEs), and no cardiovascular safety signals were identified[2]
- The results presented here are interim data (cutoff date September 22, 2022) from the OLE of X-TOLE, in which patients received open-label XEN1101 at a dose of 20 mg once daily (QD) with food

METHODS

- The study design for the X-TOLE study (ClinicalTrials.gov identifier, NCT03796962)[2] is shown in **Figure 1**

Figure 1. X-TOLE Study Design

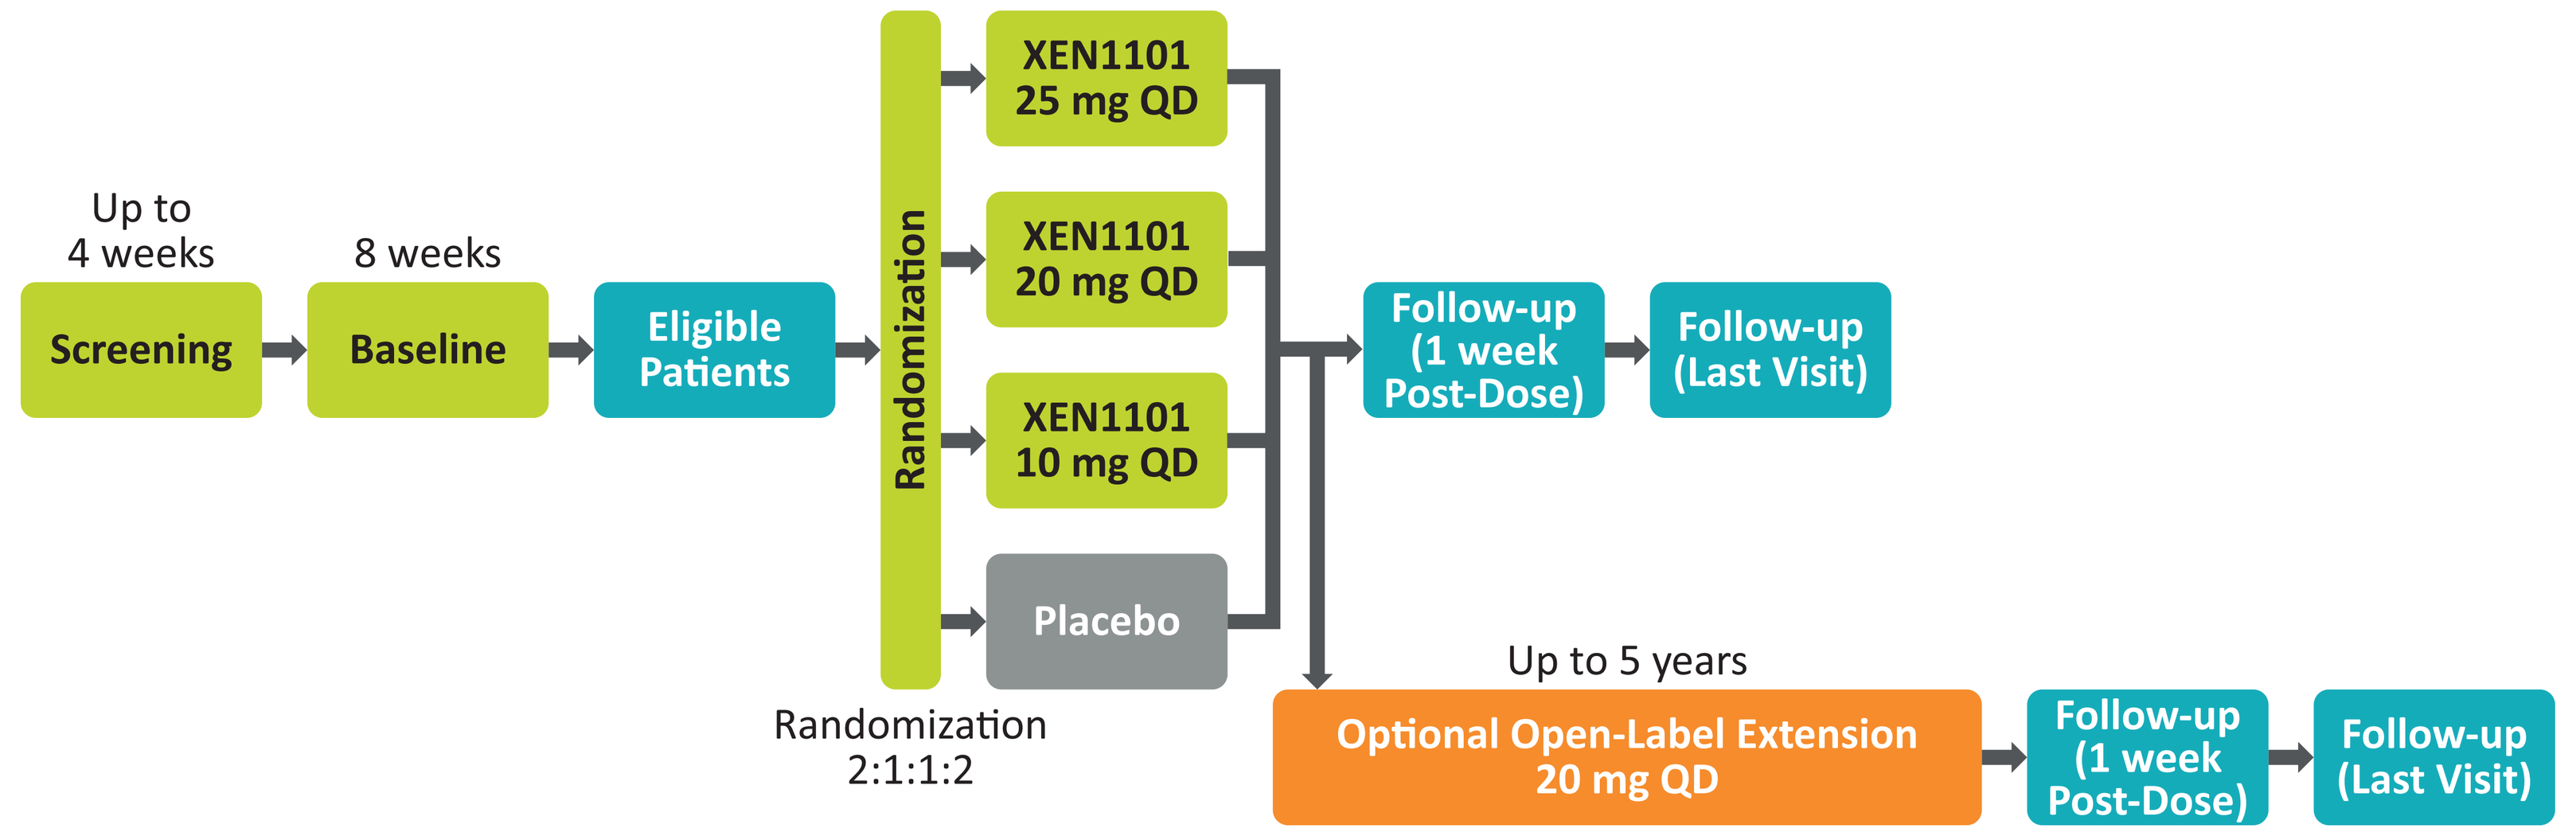

Up to 4 weeks
8 weeks
Screening
Baseline
Eligible Patients
Randomization
XEN1101 25 mg QD
XEN1101 20 mg QD
XEN1101 10 mg QD
Placebo
Randomization 2:1:1:2
Follow-up (1 week Post-Dose)
Follow-up (Last Visit)
Up to 5 years
Optional Open-Label Extension 20 mg QD
Follow-up (1 week Post-Dose)
Follow-up (Last Visit)

QD, once daily.

- The key eligibility criteria for the DBP were as follows:
 - Aged 18–75 years (inclusive) with a diagnosis of focal epilepsy per International League Against Epilepsy criteria (≥2 years)[3]
 - Receiving stable treatment with 1–3 ASMs
 - Countable seizure frequency over the 8-week baseline period of ≥4 focal seizures per month on average, recorded in an eDiary
- Patients who successfully completed the DBP with a minimum of 80% compliance with the study medication were eligible to enroll in the OLE
- Patients enrolled in the OLE received XEN1101 20 mg QD taken with the evening meal
- Efficacy in the OLE was evaluated by median percent change (MPC) in monthly FOS frequency from DBP baseline and percentage of patients with ≥50% reduction from DBP baseline in monthly FOS frequency
- Safety was assessed as severity and frequency of treatment-emergent adverse events (TEAEs) and SAEs, clinically significant changes in laboratory findings, and other measures
- Assessments occurred at week 3 in the OLE (study day 77, week 11 from randomization) and 3-month intervals thereafter for the first year
- After the first year, on-site visits occurred at 6-month intervals with teleconferences at 3 months between each on-site visit

RESULTS

Patients

- Of the 285 patients who completed the DBP, 275 (96.5%) enrolled in the OLE
- Demographics and baseline characteristics of patients in the OLE were consistent with those observed in the DBP (**Table 1**)

Table 1. Demographics and Baseline* Characteristics of the OLE Population

	OLE Population (N=275)
Age at study entry, mean (SD), y	41.1 (13.3)
Sex, n (%)	
Male	137 (49.8)
Female	138 (50.2)
Race, n (%)	
White	250 (90.9)
Black	11 (4.0)
Other	14 (5.1)
Region, n (%)	
North America	109 (39.6)
Europe	166 (60.4)
BMI, kg/m^2, mean (SD)	27.0 (5.2)
Age at epilepsy onset, mean (SD), y	18.1 (13.8)
Baseline seizure rate per mo, median (IQR)	13.5 (7.9, 30.3)
Number of prestudy ASMs failed, mean (SD)	6.5 (3.68)
Background ASM use, n (%)	
1 ASM	23 (8.4)
2 ASMs	108 (39.3)
3 ASMs	144 (52.4)
CYP3A4 inducer use, n (%)	160 (58.2)

**DBP baseline. ASM, antiseizure medication; BMI, body mass index; CYP3A4, cytochrome P450 3A4; DBP, double-blind period; IQR, interquartile range; OLE, open-label extension.*

- At the analysis cutoff (September 22, 2022), 168 patients continued to participate in the OLE
 - The most common reasons for discontinuation were lack of efficacy (12.7%), adverse events (AEs, 10.5%), and study withdrawal by the patient (9.5%)
- A total of 188 patients had been treated in the OLE for ≥12 months; 111 patients had been treated for ≥18 months at the time of the analysis cutoff
- The percentage of patients continuing XEN1101 at 6 months and 12 months into the OLE study period were 76% and 68%, respectively

Efficacy

- In patients treated with XEN1101 20 mg QD during the OLE, monthly FOS reduction was maintained with 80%–90% MPC from DBP baseline observed during study months 14–20 (**Figure 2**)
- Higher reductions were observed for patients who were receiving 1–2 ASMs at baseline compared with those receiving 3 ASMs (**Figure 3**)
- 10.5% of patients (29/275) achieved seizure freedom for any consecutive ≥12-month duration, and 17.5% (48/275) were seizure free for any ≥6 consecutive months. Responder rates are summarized in **Figure 4**

Figure 2. MPC in Monthly FOS Frequency During DBP and OLE

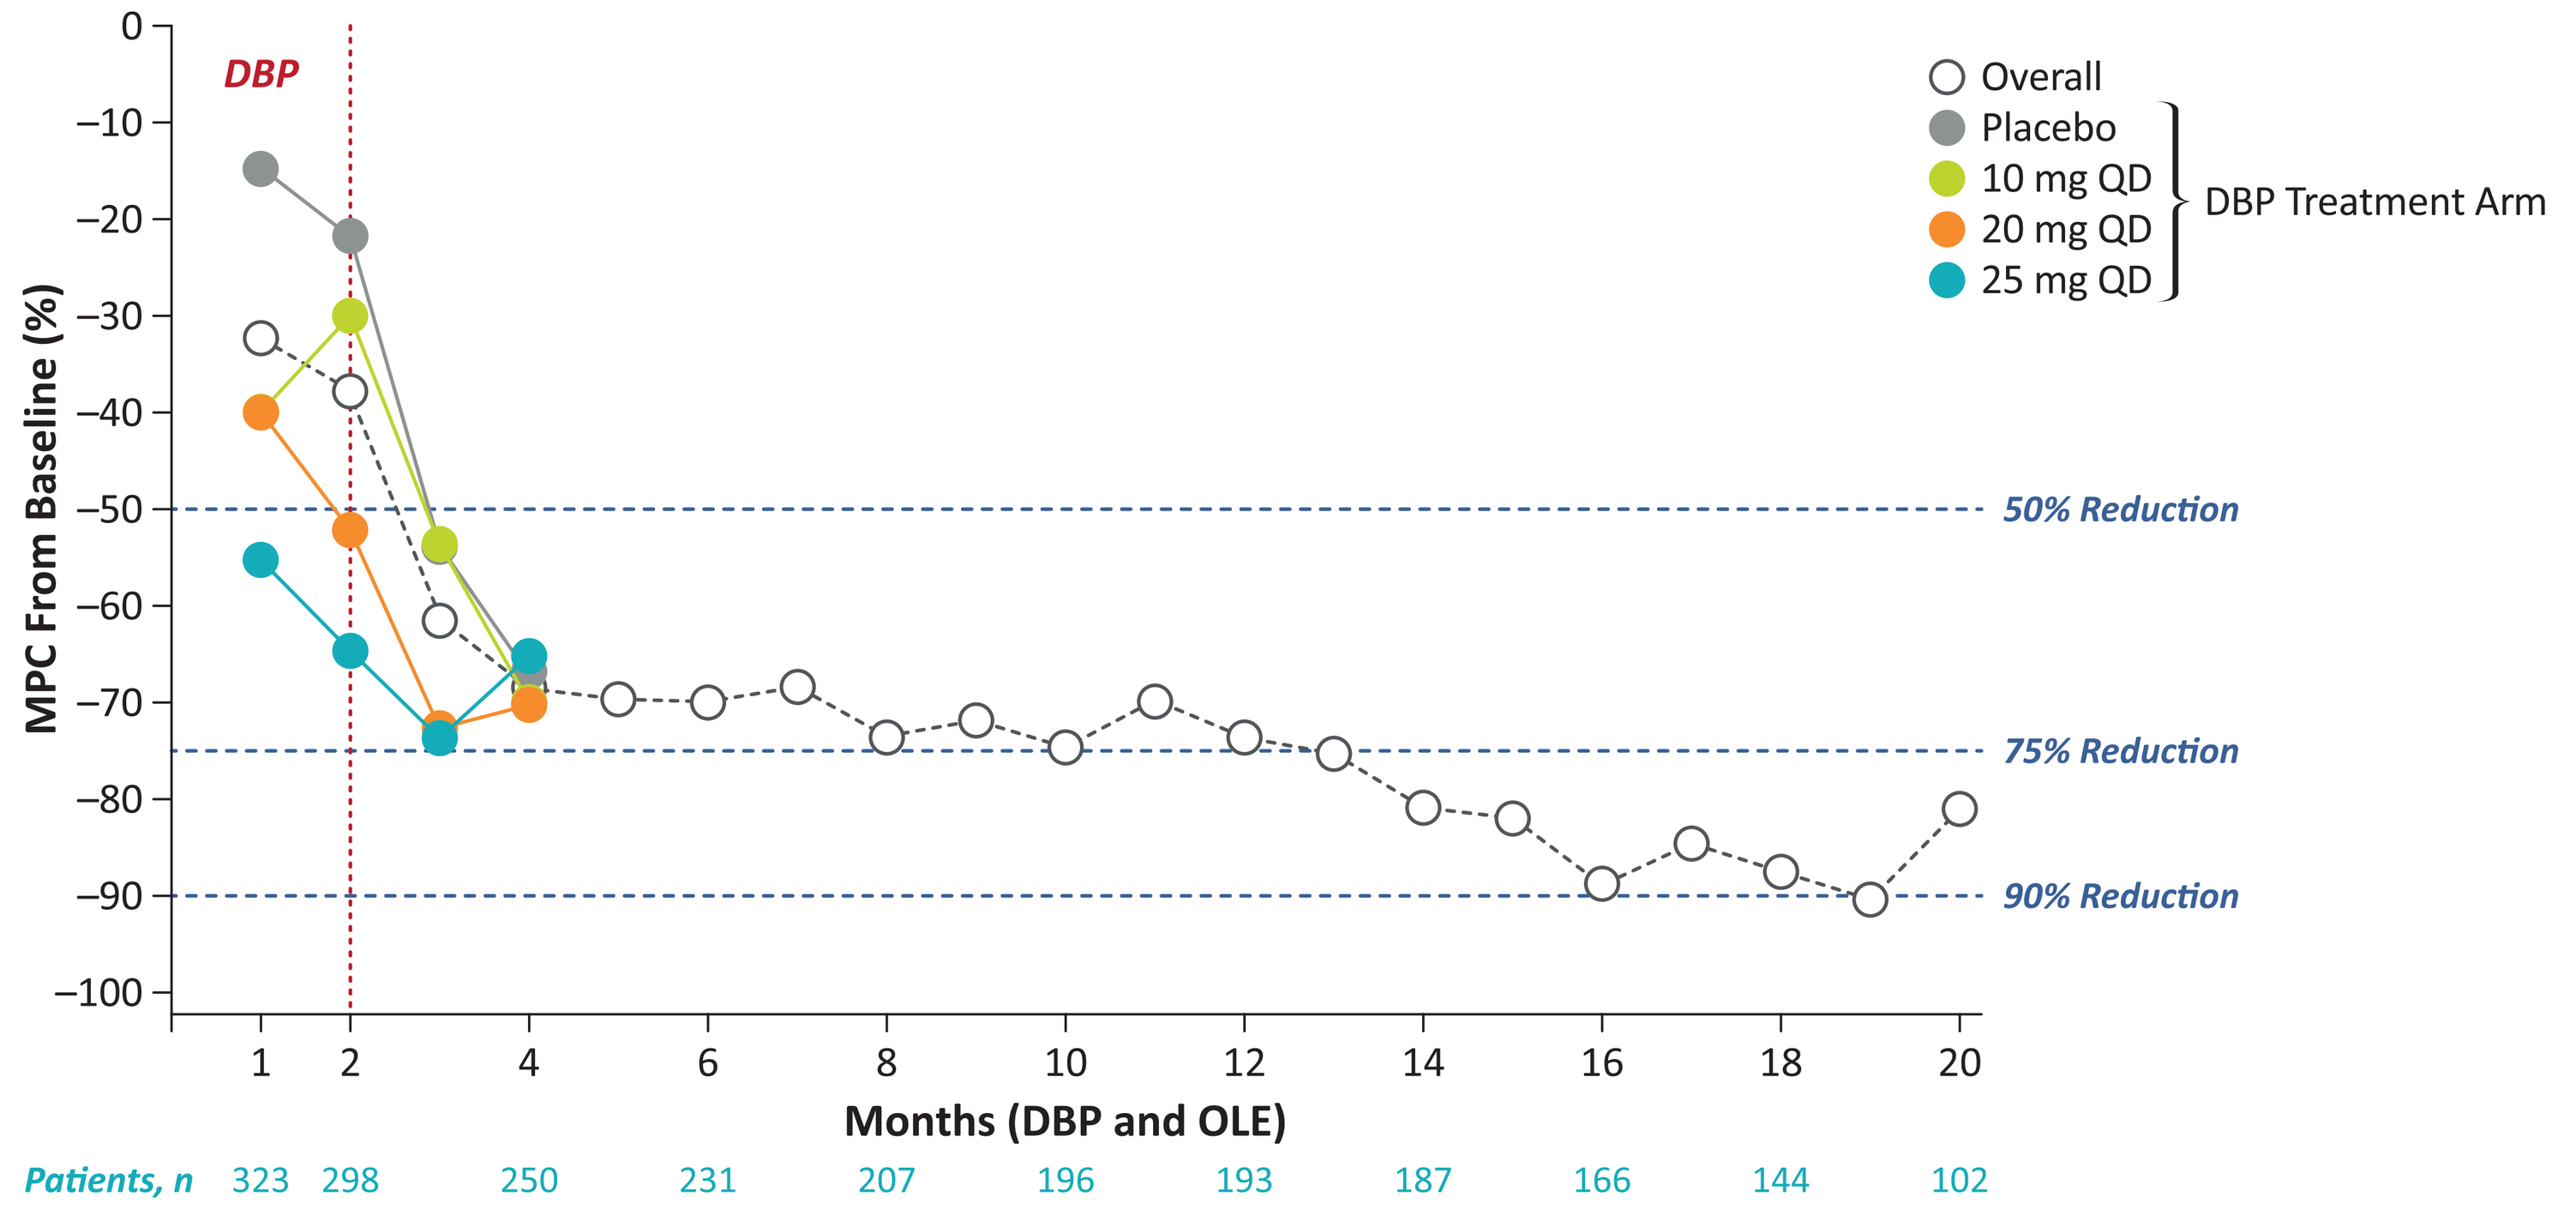

DBP
Overall
Placebo
10 mg QD
20 mg QD
25 mg QD
DBP Treatment Arm
50% Reduction
75% Reduction
90% Reduction
MPC From Baseline (%)
Months (DBP and OLE)
Patients, n 323 298 250 231 207 196 193 187 166 144 102

Note: Following DBP, all patients received 20 mg QD at start of OLE (at month 2); OLE patients separated by prior DBP treatment groups shown for first 2 months of OLE (months 3–4). DBP, double-blind period; FOS, focal onset seizures; MPC, median percentage change; OLE, open-label extension; QD, once daily.

Figure 3. MPC in Monthly FOS Frequency During DBP and OLE by Baseline Number of ASMs

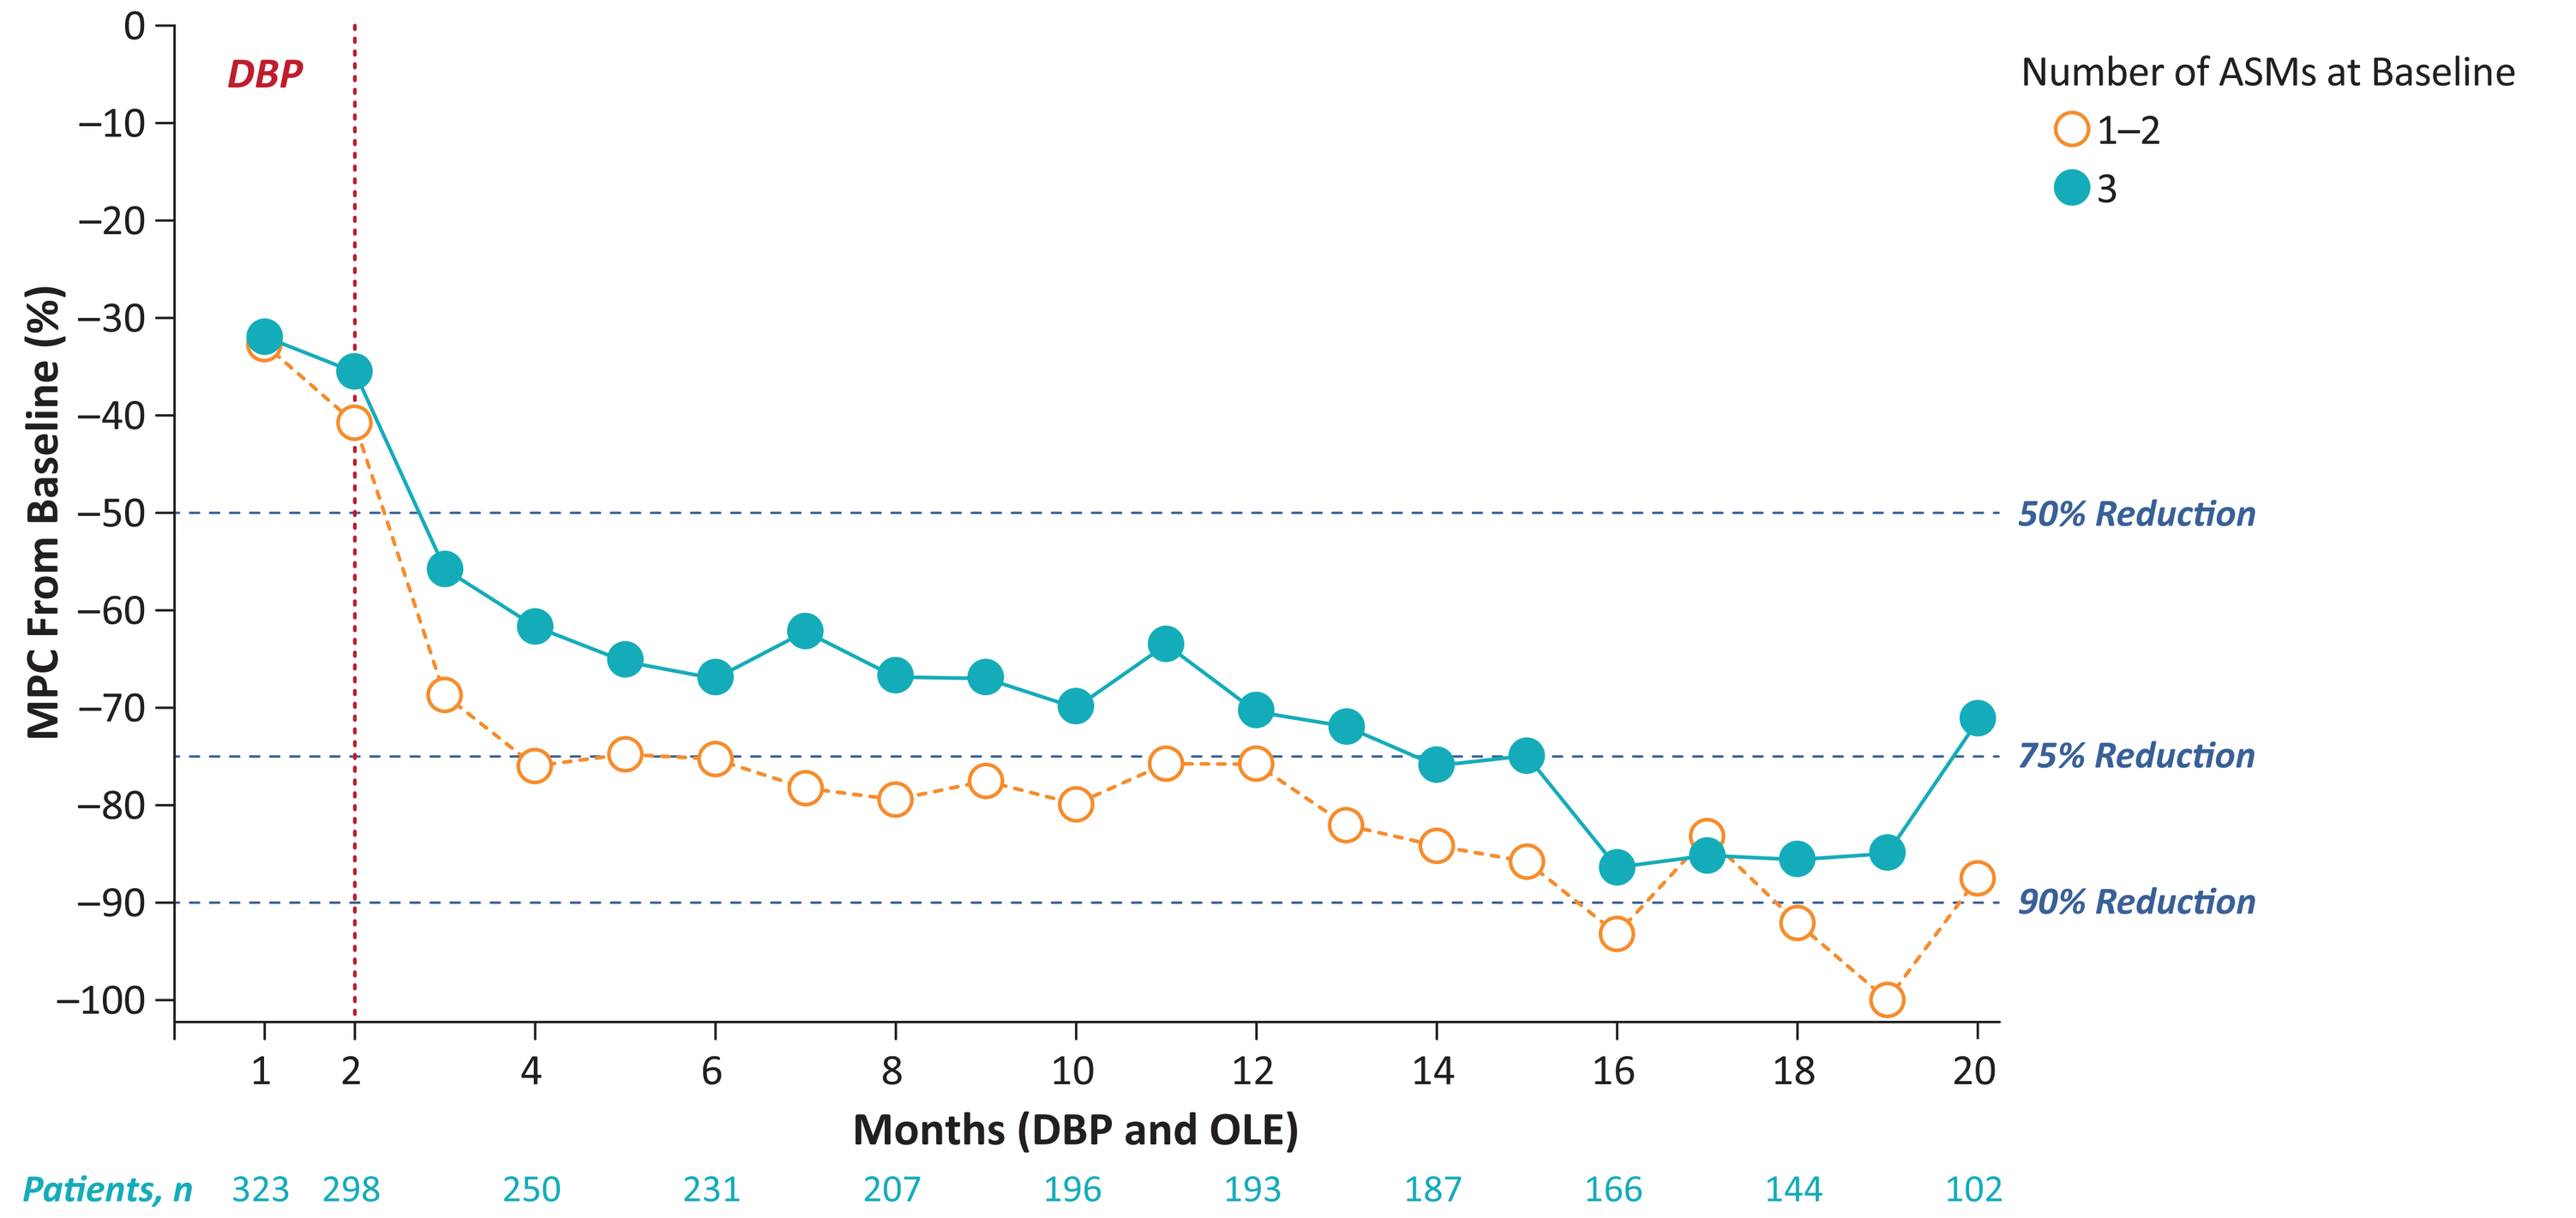

DBP
Number of ASMs at Baseline
1–2
3
50% Reduction
75% Reduction
90% Reduction
MPC From Baseline (%)
Months (DBP and OLE)
Patients, n 323 298 250 231 207 196 193 187 166 144 102

ASM, antiseizure medication; DBP, double-blind period; FOS, focal onset seizures; MPC, median percentage change; OLE, open-label extension.

Figure 4. Fraction of Patients* Maintaining Specific Levels of Monthly Median Percentage Seizure Reduction From Baseline for Consecutive Periods of ≥6, ≥12, and ≥18 Months During the OLE

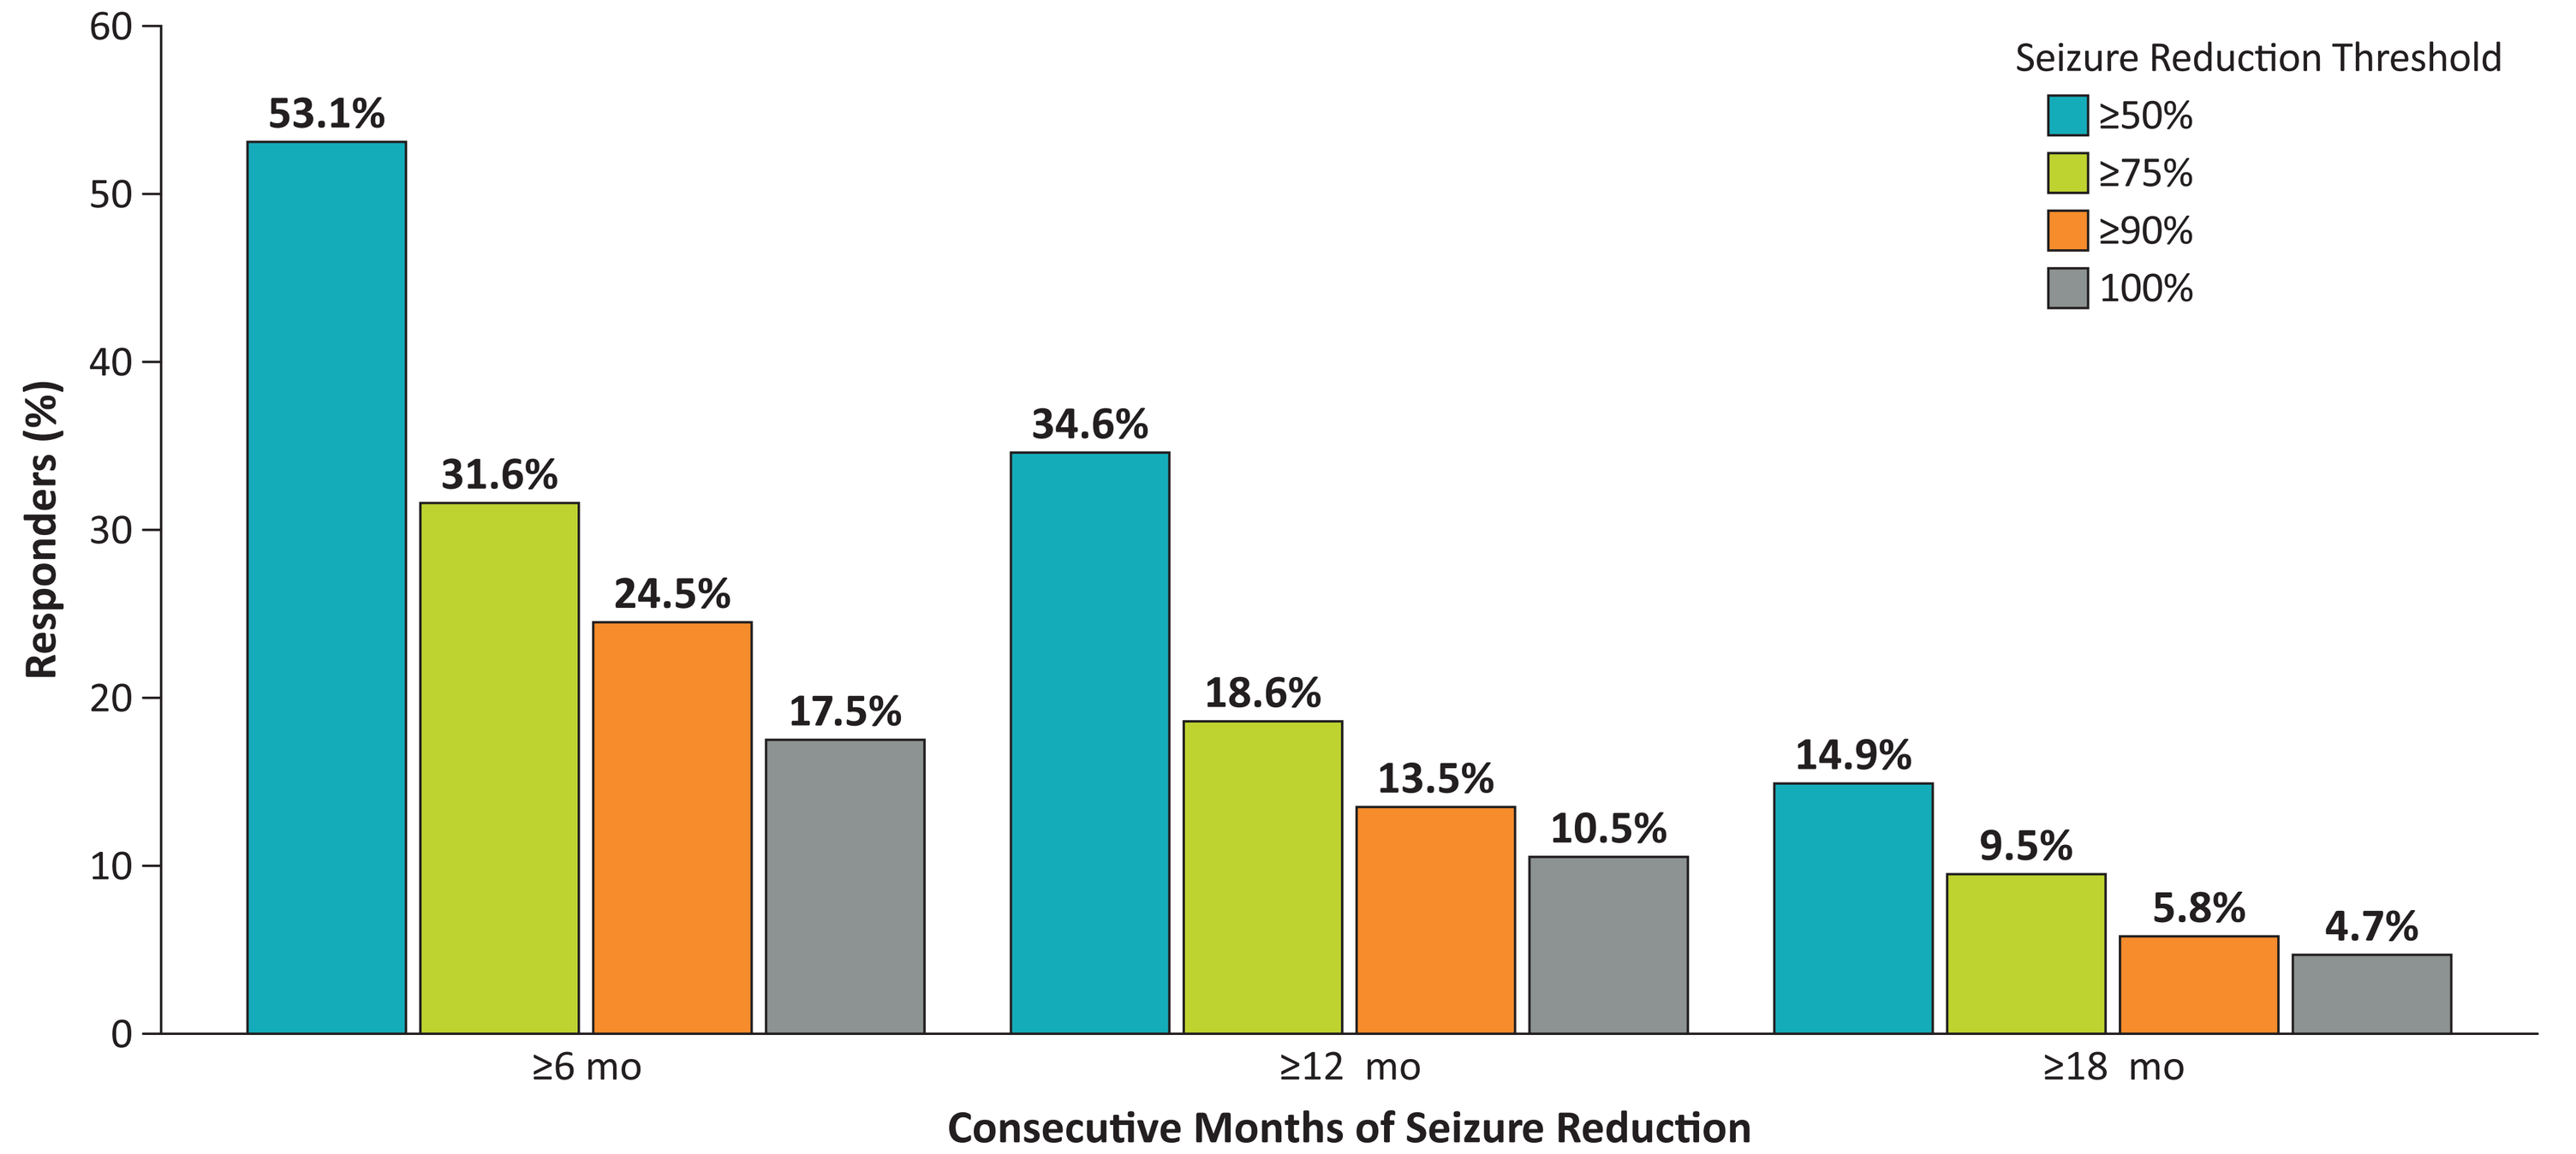

Seizure Reduction Threshold
≥50%
≥75%
≥90%
100%
Responders (%)
53.1%
31.6%
24.5%
17.5%
34.6%
18.6%
13.5%
10.5%
14.9%
9.5%
5.8%
4.7%
≥6 mo
≥12 mo
≥18 mo
Consecutive Months of Seizure Reduction

**All patients who entered the OLE (n=275). Not all patients have reached 18 months in OLE as of data cut-off. OLE, open-label extension.*

Safety

- XEN1101 20 mg QD was generally well tolerated, and the safety profile observed was similar to that of the DBP. No new safety signals were identified
- At the end of the first year patients recorded a mean (SD) weight gain of 1.1 (5.9) kg
- TEAEs occurred in 85.5% of the safety population; the most common TEAEs are summarized in **Table 2**

Table 2. TEAEs During OLE Period

Summary of TEAEs, n (%)	XEN1101 20 mg (n=275)
At least 1 TEAE	235 (85.5)
At least 1 serious TEAE	26 (9.5)
At least 1 TEAE leading to permanent treatment discontinuation	31 (11.3)
At least 1 serious TEAE leading to death	1 (0.4)
Most common AEs (≥5% of overall OLE population), n (%)	
Dizziness	57 (20.7)
Headache	37 (13.5)
Coronavirus infection	32 (11.6)
Fall	31 (11.3)
Somnolence	27 (9.8)
Weight increased	25 (9.1)
Gait disturbance	24 (8.7)
Fatigue	20 (7.3)
Aphasia	19 (6.9)
Urinary tract infection	18 (6.5)
Memory impairment	17 (6.2)
Confusional state	15 (5.5)
Disturbance in attention	14 (5.1)
Tremor	14 (5.1)

AE, adverse event; OLE, open-label extension; TEAE, treatment-emergent adverse event.

- In addition to the TEAEs summarized in **Table 2**, 2 patients reported urinary retention, 1 reported as mild and the other moderate; no dose changes were made in either case
- As shown in **Table 2**, SAEs were reported in 26 (9.5%) patients. The only SAEs reported in >1 patients were seizures in 5 (1.8%) patients, and paresthesia and deep vein thrombosis reported in 2 (0.7%) patients
- There was 1 sudden unexplained death in epilepsy reported, determined by the investigator not to be related to the study drug

CONCLUSIONS

- XEN1101 20 mg QD yielded long-term efficacy in this interim analysis with 68% retention at 12 months
- During study months 14–20, there was a sustained monthly reduction in seizure frequency (80%–90% MPC) from DBP baseline
- Seizure freedom for ≥6-month and ≥12-month consecutive durations was achieved in 17.5% and 10.5% of patients, respectively
- XEN1101 continues to be generally well-tolerated in the OLE with AEs consistent with prior results and other ASMs; no new safety signals were identified

ACKNOWLEDGMENTS Medical writing support was provided by Valerie Kinchen, PhD, of The Curry Rockefeller Group, LLC (Tarrytown, NY), and was funded by Xenon Pharmaceuticals Inc.

FUNDING This study was funded by Xenon Pharmaceuticals Inc.

DISCLOSURES Jacqueline French receives salary support from the Epilepsy Foundation and for consulting work and/or attending scientific advisory boards on behalf of the Epilepsy Study Consortium for Aeonian/Aeovian; Alterity Therapeutics Ltd.; Anavex; Arkin Holdings; Angelini Pharma S.p.A; Arvelle Therapeutics, Inc.; Athenen Therapeutics/Carnot Pharma; Autifony Therapeutics Ltd.; Baergic Bio; Beacon Biosignals, Inc.; Biogen; Biohaven Pharmaceuticals; BioMarin Pharmaceutical, Inc.; BioXcel Therapeutics; Bloom Science, Inc.; BridgeBio Pharma, Inc.; Camp4 Therapeutics Corp.; Cerebral Therapeutics; Cerevel; Clinical Education Alliance; Coda Biotherapeutics; Corlieve Therapeutics; Crossject; Eisai; Eliem Therapeutics; Encoded Therapeutics; Engage Therapeutics; Engrail; Epalex; Epihunter; Epiminder; Epitel, Inc.; Equilibre BioPharmaceuticals; Greenwich Biosciences; Grin Therapeutics; GW Pharma; Janssen Pharmaceutica; Jazz Pharmaceuticals; Knopp Biosciences; Korro Bio, Inc.; Lipocine; LivaNova; Longboard Pharmaceuticals; Lundbeck; Marinus; Mend Neuroscience; Merck; NeuCyte, Inc.; Neumirna Therapeutics; Neurocrine; Neuroelectrics USA Corp.; Neuronetics, Inc.; Neuropace; NxGen Medicine, Inc.; Ono Pharmaceutical Co.; Otsuka Pharmaceutical Development; Ovid Therapeutics, Inc.; Paladin Labs, Inc.; Passage Bio; Pfizer; Praxis; PureTech LTY, Inc.; Rafa Laboratories Ltd.; Receptor Holdings, Inc.; SK Life Science; Sofinnova; Stoke; Supernus; Synergia Medical; Takeda; Third Rock Ventures LLC; UCB, Inc.; Ventus Therapeutics; Xenon Pharmaceuticals Inc.; Xeris; Zogenix; and Zynerba. She has received research support from the Epilepsy Study Consortium (funded by Andrews Foundation, Eisai, Engage, Lundbeck, Pfizer, SK Life Science, Sunovion, UCB, Vogelstein Foundation), Epilepsy Study Consortium/Epilepsy Foundation (funded by UCB), GW/FACES, and National Institute of Neurological Disorders and Stroke. She has served on the editorial boards for Lancet Neurology and Neurology Today and as Chief Medical/Innovation Officer for the Epilepsy Foundation. She has received travel reimbursement related to research, advisory meetings, or presentation of results at scientific meetings from the Epilepsy Study Consortium; Epilepsy Foundation; Angelini Pharma S.p.A.; Cerevel; Clinical Education Alliance; NeuCyte, Inc.; Neurocrine; Praxis; and Xenon Pharmaceuticals Inc. Roger Porter is a consultant for Aeterna, Cadent, Engrail, Longboard, Neurocrine, Otsuka, Passage Bio, and Xenon Pharmaceuticals Inc. Emilio Perucca has received speaker or consultancy fees from Angelini, Arvelle, Biogen, Eisai, GW Pharma, Janssen, PMI Life Sciences, Sanofi, Shackelford Pharma, SK Life Science, Sun Pharma, Takeda, UCB Pharma, Xenon Pharmaceuticals Inc., and Zogenix. Martin Brodie has nothing to declare. Michael A. Rogawski is a paid consultant to Xenon Pharmaceuticals Inc. Cynthia Harden, Jenny Qian, Constanza Luzon Rosenblut, Christopher Kenney, and Gregory N. Beatch are employees of and own stock or stock options in Xenon Pharmaceuticals Inc.

REFERENCES 1. Chen Z, et al. *JAMA Neurol.* 2018;75(3):279-286. 2. French J, et al. *Neurology.* 2022;98(18 SUPPL). 3. Fisher RS, et al. *Epilepsia.* 2017;58(4):522-530.

Poster No. 2.235 | American Epilepsy Society Annual Meeting | Nashville, TN | December 2–6, 2022

XENON