Use of Transcranial Magnetic Stimulation Data in the Design of a Dose Ranging Finding Efficacy, Safety, Tolerability and Pharmacokinetics Study of XEN1101 in Patients with Focal Epilepsy

Greg N. Beatch[1], Cynthia Harden[1], Rostam Namdari[1], Jay Cadieux[1], Ron Ablog[1], Heather Kato[1], Dorothea Scholl[1], Lynn Murray[1], Steven Evans[1], Simon N. Pimstone[1], Robin Sherrington[1], Charles J. Cohen[1], James Empfield[1], Christopher Crean[2], Darby Thompson[3], Ernesto Aycardi[1]

[1]*Xenon Pharmaceuticals Inc. 3650 Gilmore Way, Burnaby, BC, Canada;* [2] *Xyzagen Inc. 480 Hillsboro Street, Suite 140, Pittsboro, NC, USA;* [3] *EMMES Canada 200-4664 Lougheed Hwy, Burnaby, BC, Canada*

BACKGROUND

XEN1101: A differentiated K_V7 potassium channel modulator being developed for the treatment of epilepsy

- XEN1101 is an investigational drug that enhances activation of neuronal $K_V7.2$-7.5 (KCNQ2-5) potassium channels. XEN1101 is currently in Phase 2 clinical development by Xenon Pharmaceuticals for the treatment of epilepsy.
- XEN1101 is significantly differentiated from the first generation $K_V7.2$-7.5 opener, ezogabine, with higher *in vitro* and *in vivo* potency and improved pharmacokinetics (PK) supporting once a day dosing.
- The objectives of the ongoing Phase 2 clinical study are to evaluate the efficacy, safety, tolerability and PK of oral XEN1101 in patients with refractory focal seizures. The doses for this study were selected based on XEN1101's side effect profile in Phase 1 studies and surrogate pharmacodynamic endpoints obtained from transcranial magnetic stimulation (TMS) studies conducted in healthy adult subjects.

PHASE 1 RESULTS

Table 1: Dose-Related Adverse Events (AEs) Occurring in ≥2 Subjects Overall for Multiple Ascending Dose Cohorts in First-in-Human Phase 1 Study

System Organ Class / Preferred Term	XEN1101 Cohort 1 15 mg QD (fasted) (N=6)	XEN1101 Cohort 2 15 mg QD (fed) (N=6)	XEN1101 Cohort 3 25 mg QD (fed) (N=6)	XEN1101 Overall (N=18)	Placebo Pooled (N=6)
	n (%) E	n (%) E	n (%) E	n (%) E	n (%) E
Subjects with at least one TEAE	4 (66.7) 11	4 (66.7) 18	6 (100.0) 39	14 (77.8) 68	2 (33.3) 5
Cardiac Disorders	0	2 (33.3) 2	0	2 (11.1) 2	0
Palpitations	0	2 (33.3) 2	0	2 (11.1) 2	0
Eye Disorders	1 (16.7) 1	0	5 (83.3) 5	6 (33.3) 6	0
Vision blurred	0	0	5 (83.3) 5	5 (27.8) 5	0
Musculoskeletal and Connective Tissue Disorders	0	0	2 (33.3) 3	2 (11.1) 3	1 (16.7) 1
Muscle twitching	0	0	2 (33.3) 2	2 (11.1) 2	1 (16.7) 1
Nervous System Disorders	3 (50.0) 6	3 (50.0) 13	6 (100.0) 23	12 (66.7) 42	2 (33.3) 4
Balance disorder	1 (16.7) 1	1 (16.7) 1	1 (16.7) 1	3 (16.7) 3	0
Dizziness	0	1 (16.7) 1	2 (33.3) 2	3 (16.7) 3	0
Headache	1 (16.7) 1	3 (50.0) 3	3 (50.0) 4	7 (38.9) 8	0
Memory impairment	2 (33.3) 2	1 (16.7) 1	2 (33.3) 2	5 (27.8) 5	0
Sensory disturbance	0	0	2 (33.3) 2	2 (11.1) 2	0
Somnolence	0	3 (50.0) 3	4 (66.7) 4	7 (38.9) 7	*1 (16.7) 1
Speech disorder	0	2 (33.3) 2	4 (66.7) 4	6 (33.3) 6	0
Vascular Disorders	0	1 (16.7) 1	4 (66.7) 4	5 (27.8) 5	0
Hot flush	0	1 (16.7) 1	2 (33.3) 2	3 (16.7) 3	0
Orthostatic hypotension	0	0	*2 (33.3) 2	2 (11.1) 2	0

E = number of events; n = number of subjects having an adverse event; N = Number of subjects at risk.
* Denotes moderate AEs. All other AEs were mild. There were no severe AEs, withdrawals due to AEs, or SAEs.

Relationship of XEN1101 Plasma Levels to CNS Activity

Figure 1: XEN1101-Induced Changes in Corticospinal Excitability Assessed using Transcranial Magnetic Stimulation[1]

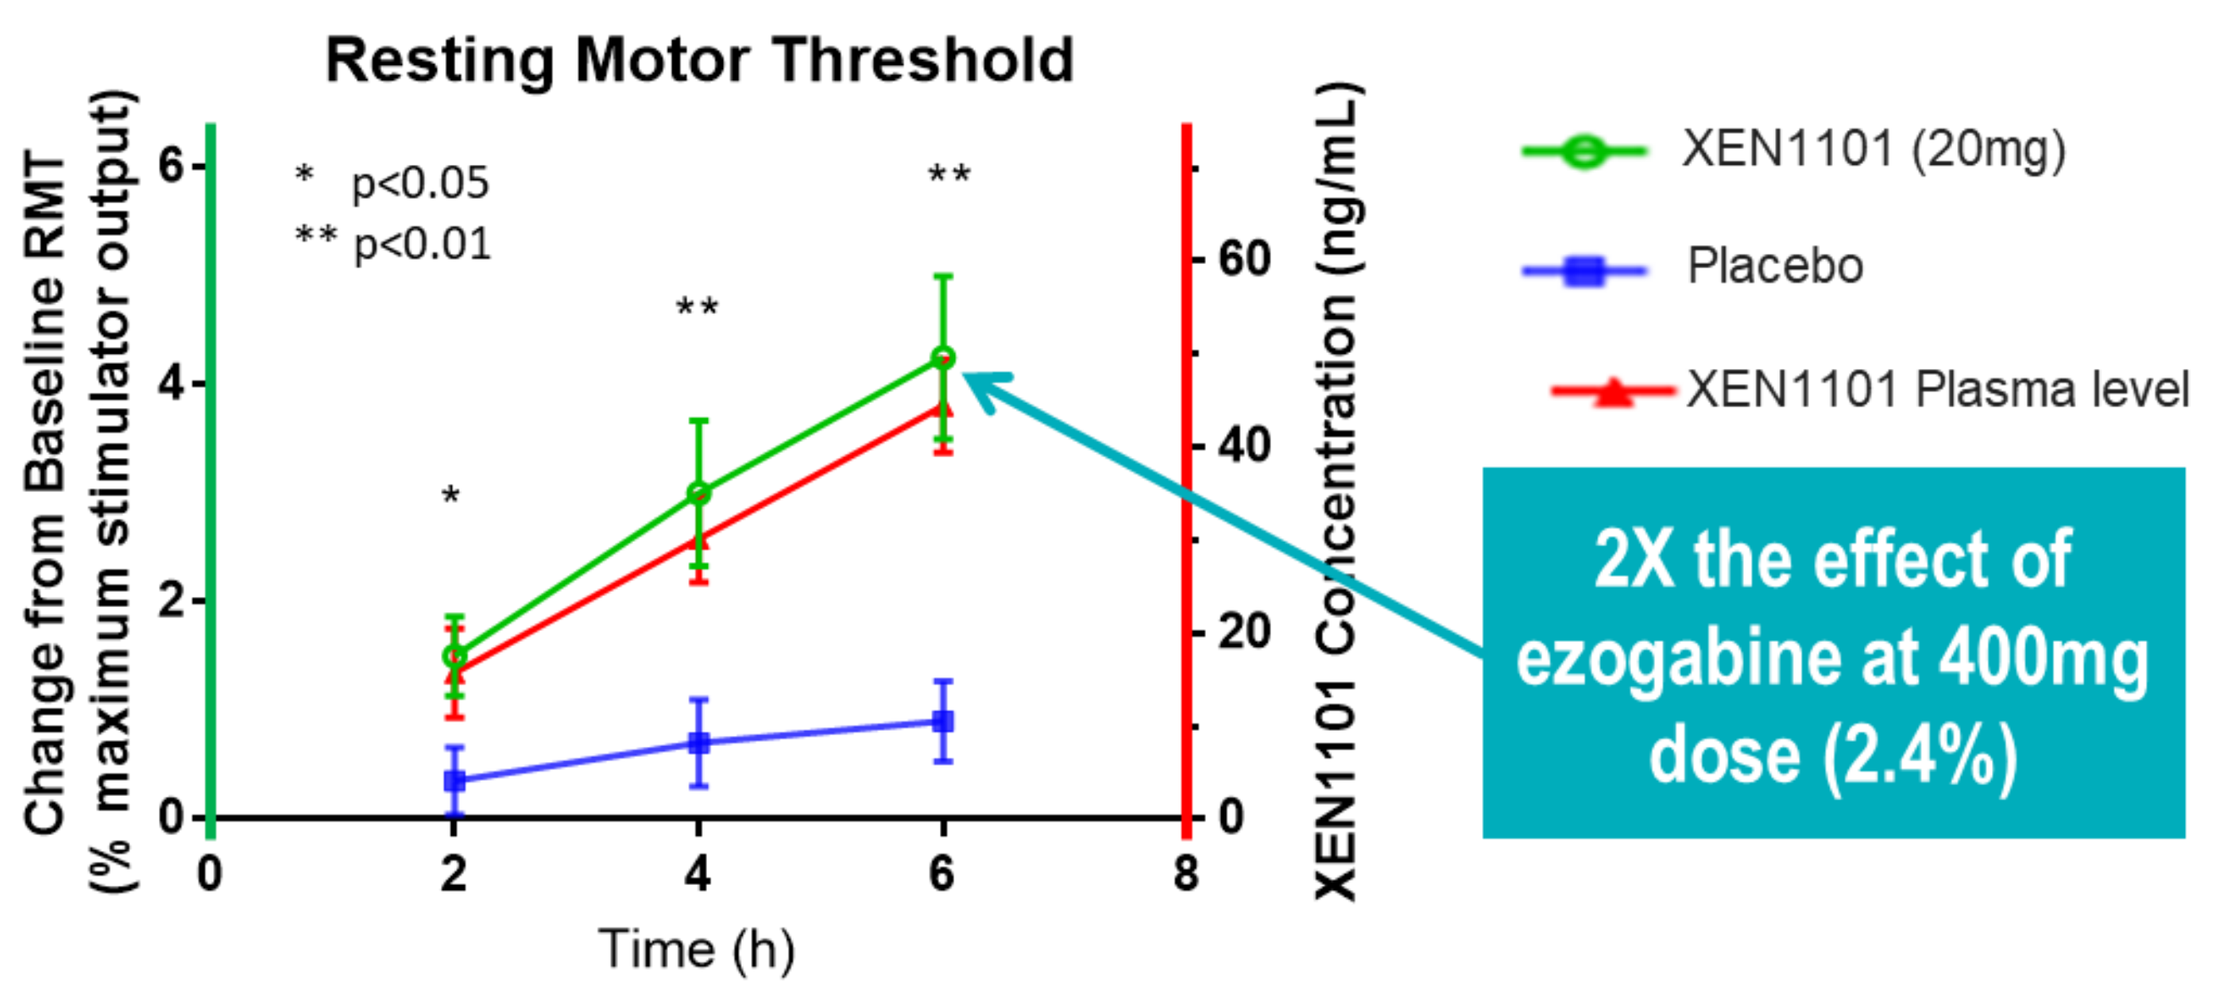
Resting Motor Threshold
Change from Baseline RMT (% maximum stimulator output)
XEN1101 Concentration (ng/mL)
Time (h)
* p<0.05
** p<0.01
XEN1101 (20mg)
Placebo
XEN1101 Plasma level
2X the effect of ezogabine at 400mg dose (2.4%)

- RMT increased in proportion to XEN1101 plasma level. Changes were significant in comparison to time matched placebo treated subjects.
- In a previously published study, ezogabine (400 mg) increased RMT by 2.4 ± 3.6% at 2 hours post dose.[2]

Use of TMS to Inform Dose Selection in Phase 2

Figure 2: Projected Plasma Levels at Doses Used in the Study (Simulations based upon PK Parameters in Phase 1)

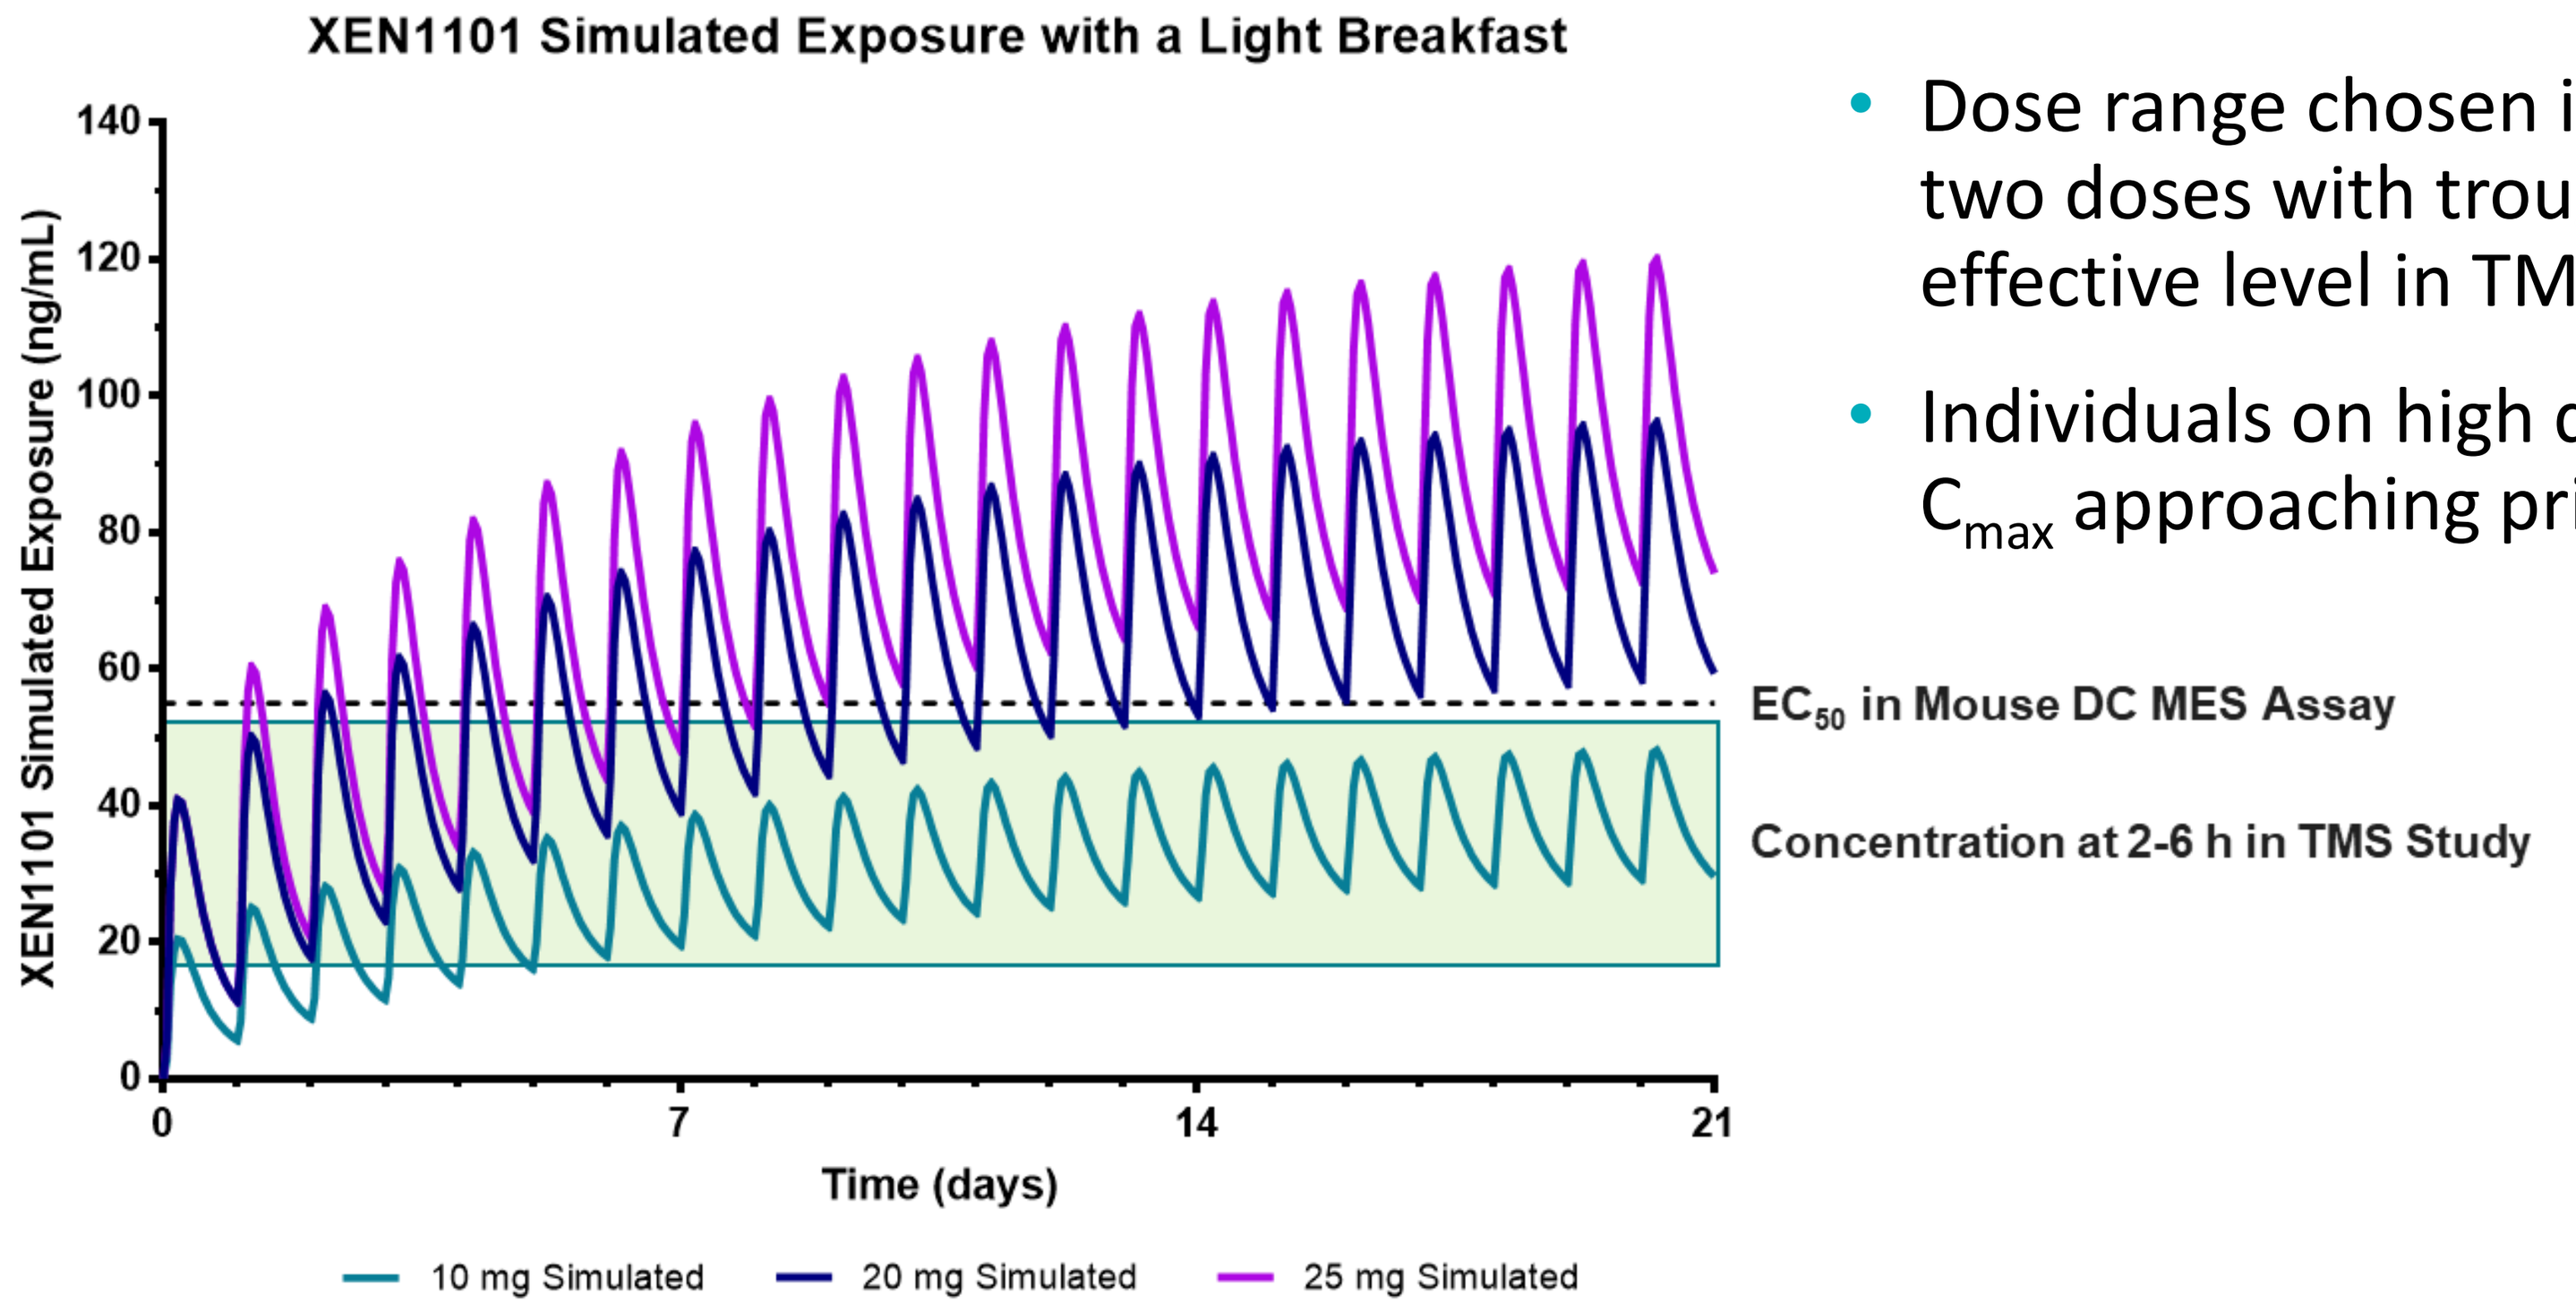
XEN1101 Simulated Exposure with a Light Breakfast
XEN1101 Simulated Exposure (ng/mL)
Time (days)
EC_{50} in Mouse DC MES Assay
Concentration at 2-6 h in TMS Study
10 mg Simulated
20 mg Simulated
25 mg Simulated

- Dose range chosen in Phase 2 will provide two doses with trough levels above effective level in TMS.
- Individuals on high dose expected to have C_{max} approaching primate NOAEL.

PHASE 2 STUDY

Endpoints and Analysis

- The primary endpoint is median percent change (MPC) in monthly (28 days) focal seizure frequency from baseline compared to the 8 week double-blind treatment period versus placebo. Including a blinded sample-size reassessment, the study will have 90% power to detect a monotonic dose response assuming a -20% MPC in placebo and -25%, -30% and -35% MPC at 10, 20 and 25 mg QD XEN1101, respectively.
- The Primary Analysis utilizes a closed, hierarchical testing procedure to control study-wide type I error to 0.05, and permits staged assessment of dose response followed by paired and grouped comparisons to placebo.
- Secondary endpoints include an evaluation of responder rate compared to placebo, as well as evaluation of changes in weekly seizure frequency and quality of life assessments.
- Exploratory endpoints include evaluation of impact of XEN1101 on the number of seizure-free days, specific seizure types, time to pre-randomization monthly focal seizure count and exploration of potential epilepsy-associated genes and biomarkers.

Study Design

- The multicenter Phase 2 clinical trial (called the X-TOLE study) is being conducted at approximately 90 sites in Europe and North America. Approximately 300 adult patients with focal epilepsy are planned to be randomized to 8 weeks treatment with XEN1101 in one of three arms (25 mg, 20 mg or 10 mg QD) or to placebo (randomized 2:1:1:2) in a double-blinded, parallel manner, stratified by background CYP inducer anti-seizure medications (ASMs).
- Significant effects on TMS measures present at 20 mg in a Phase 1 placebo-controlled trial were used to anchor the dose selection[1]. Titration at initiation of dosing, or tapering at termination of dosing, is not required due to the PK characteristics of XEN1101.
- Eligibility criteria include ≥4 countable focal seizures per month recorded with an eDiary during an 8 week baseline period, while receiving stable treatment with 1-3 ASMs. Treatment with implanted neurostimulators and/or cannabinoids is permitted, as well as benzodiazepines as rescue medications for seizure clustering.
- Plasma PK samples will be collected at each visit. Safety evaluations include adverse event (AE) monitoring, clinical laboratory tests, vital signs, ECGs, physical examinations, and Columbia-Suicide Severity Rating Scale assessment.

Figure 3: Overview of Study Design

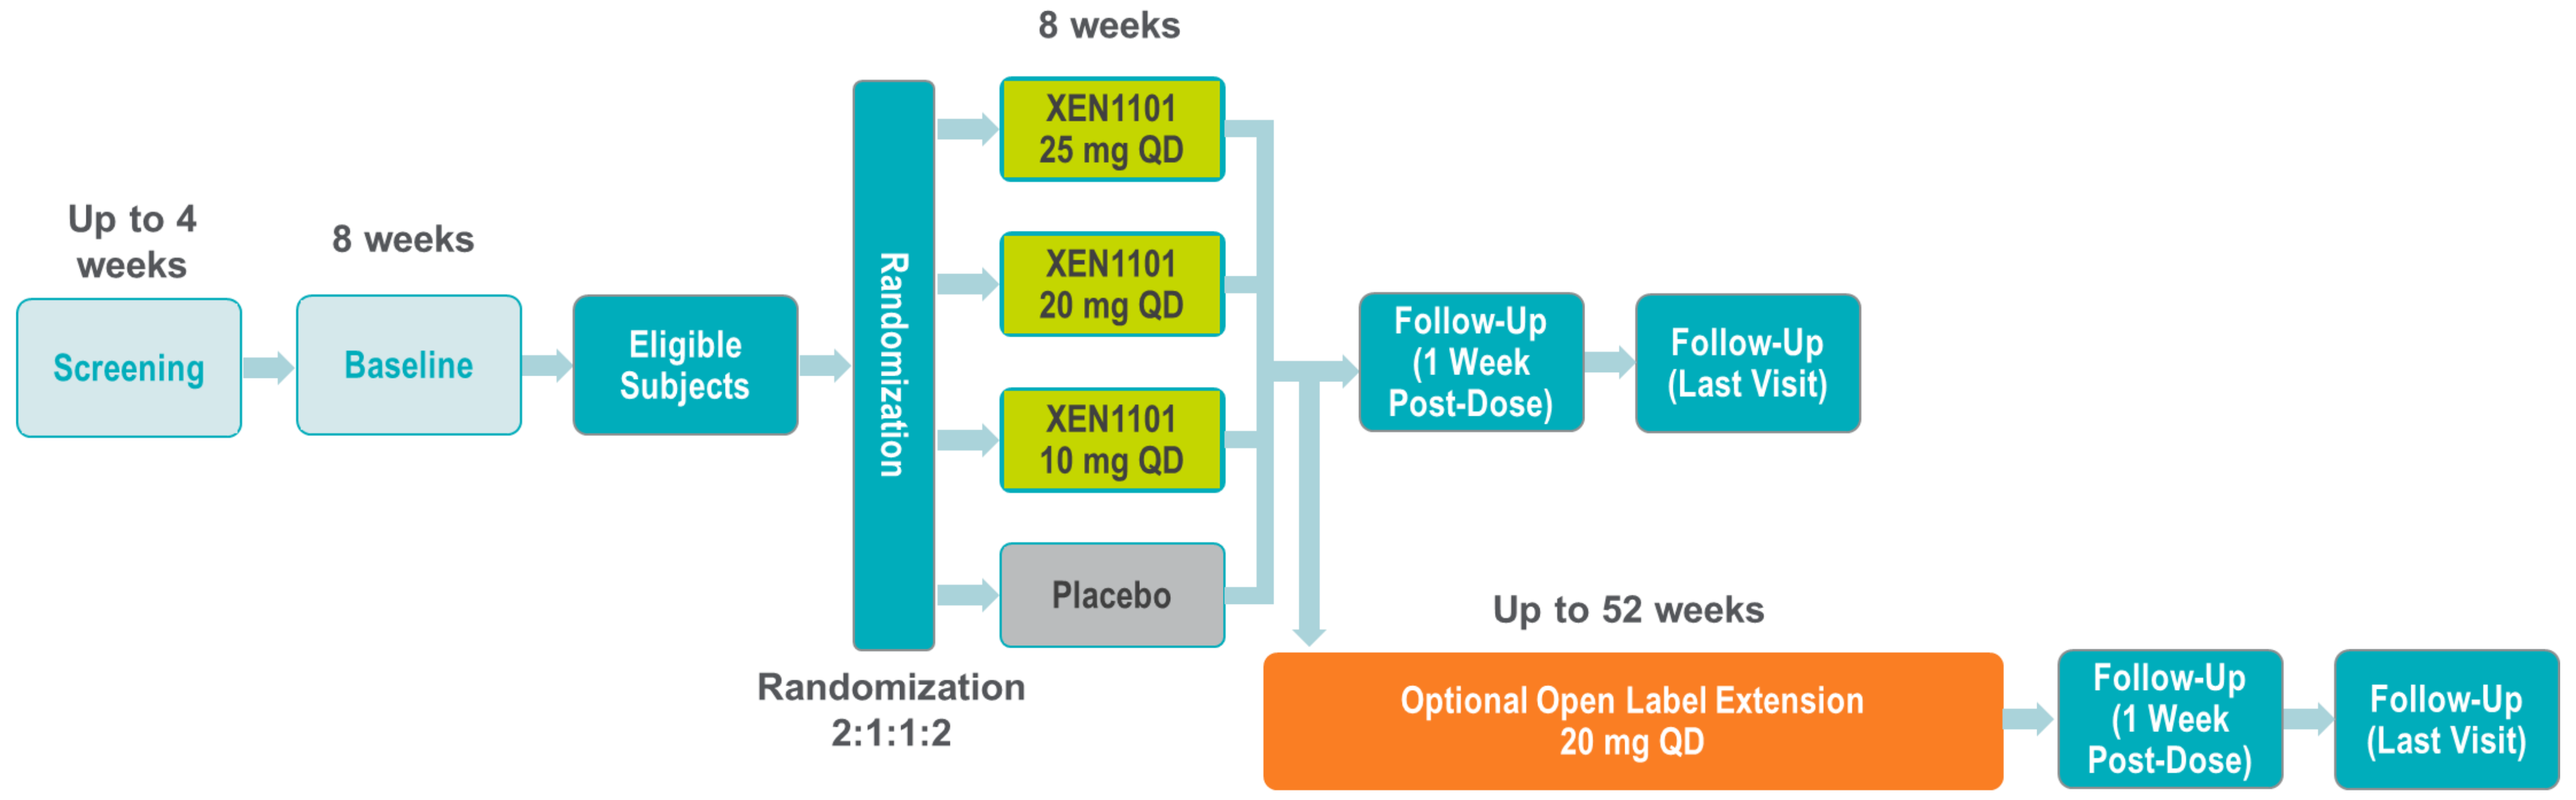
Up to 4 weeks
Screening
8 weeks
Baseline
Eligible Subjects
Randomization
8 weeks
XEN1101 25 mg QD
XEN1101 20 mg QD
XEN1101 10 mg QD
Placebo
Randomization 2:1:1:2
Follow-Up (1 Week Post-Dose)
Follow-Up (Last Visit)
Up to 52 weeks
Optional Open Label Extension 20 mg QD
Follow-Up (1 Week Post-Dose)
Follow-Up (Last Visit)

CONCLUSIONS

- The RMT signal observed with 20 mg XEN1101 in TMS is markedly greater than that observed following a 400 mg dose of ezogabine, providing confidence in the dose selection for the Phase 2 clinical trial.
- Incorporating TMS evidence of CNS activity in Phase 1 studies may be a useful adjunct in refining dose selection for Phase 2 epilepsy studies.

Next Steps

- Patients successfully completing the double blind phase of the study may be eligible for continuation at 20 mg QD for up to 1 year in the open-label, long-term extension phase of the study.

[1]Premoli, I. *et al*. (2019), TMS as a pharmacodynamic indicator of cortical activity of a novel anti-epileptic drug, XEN1101. Ann Clin Transl Neurol, 6: 2164-2174. doi:10.1002/acn3.50896

[2]Ossemann, M. *et al*. (2016). Effect of a single dose of retigabine in cortical excitability parameters: a cross-over, double-blind placebo-controlled TMS study. Epilepsy Res. 126, 78–82. doi: 10.1016/j.eplepsyres.2016.06.004

All trademarks are the property of their respective owners.

73rd Annual Meeting of the American Epilepsy Society | December 6-10, 2019 | Baltimore, MD

XENON